Post-translationally modified antigens are good targets for cancer immunotherapy but some patients have antigen specific T-regs that may need to be neutralized

Suha Atabani [1], Victoria Brentville [2], Ian Daniels [2], Ruhul Choudhury [2], Katherine Cook [2], Poulam Patel [1] and Lindy Durrant [1] [2].

[1]Nottingham University, Nottingham, UK, [2]Scancell Holdings plc, Oxford UK

INTRODUCTION

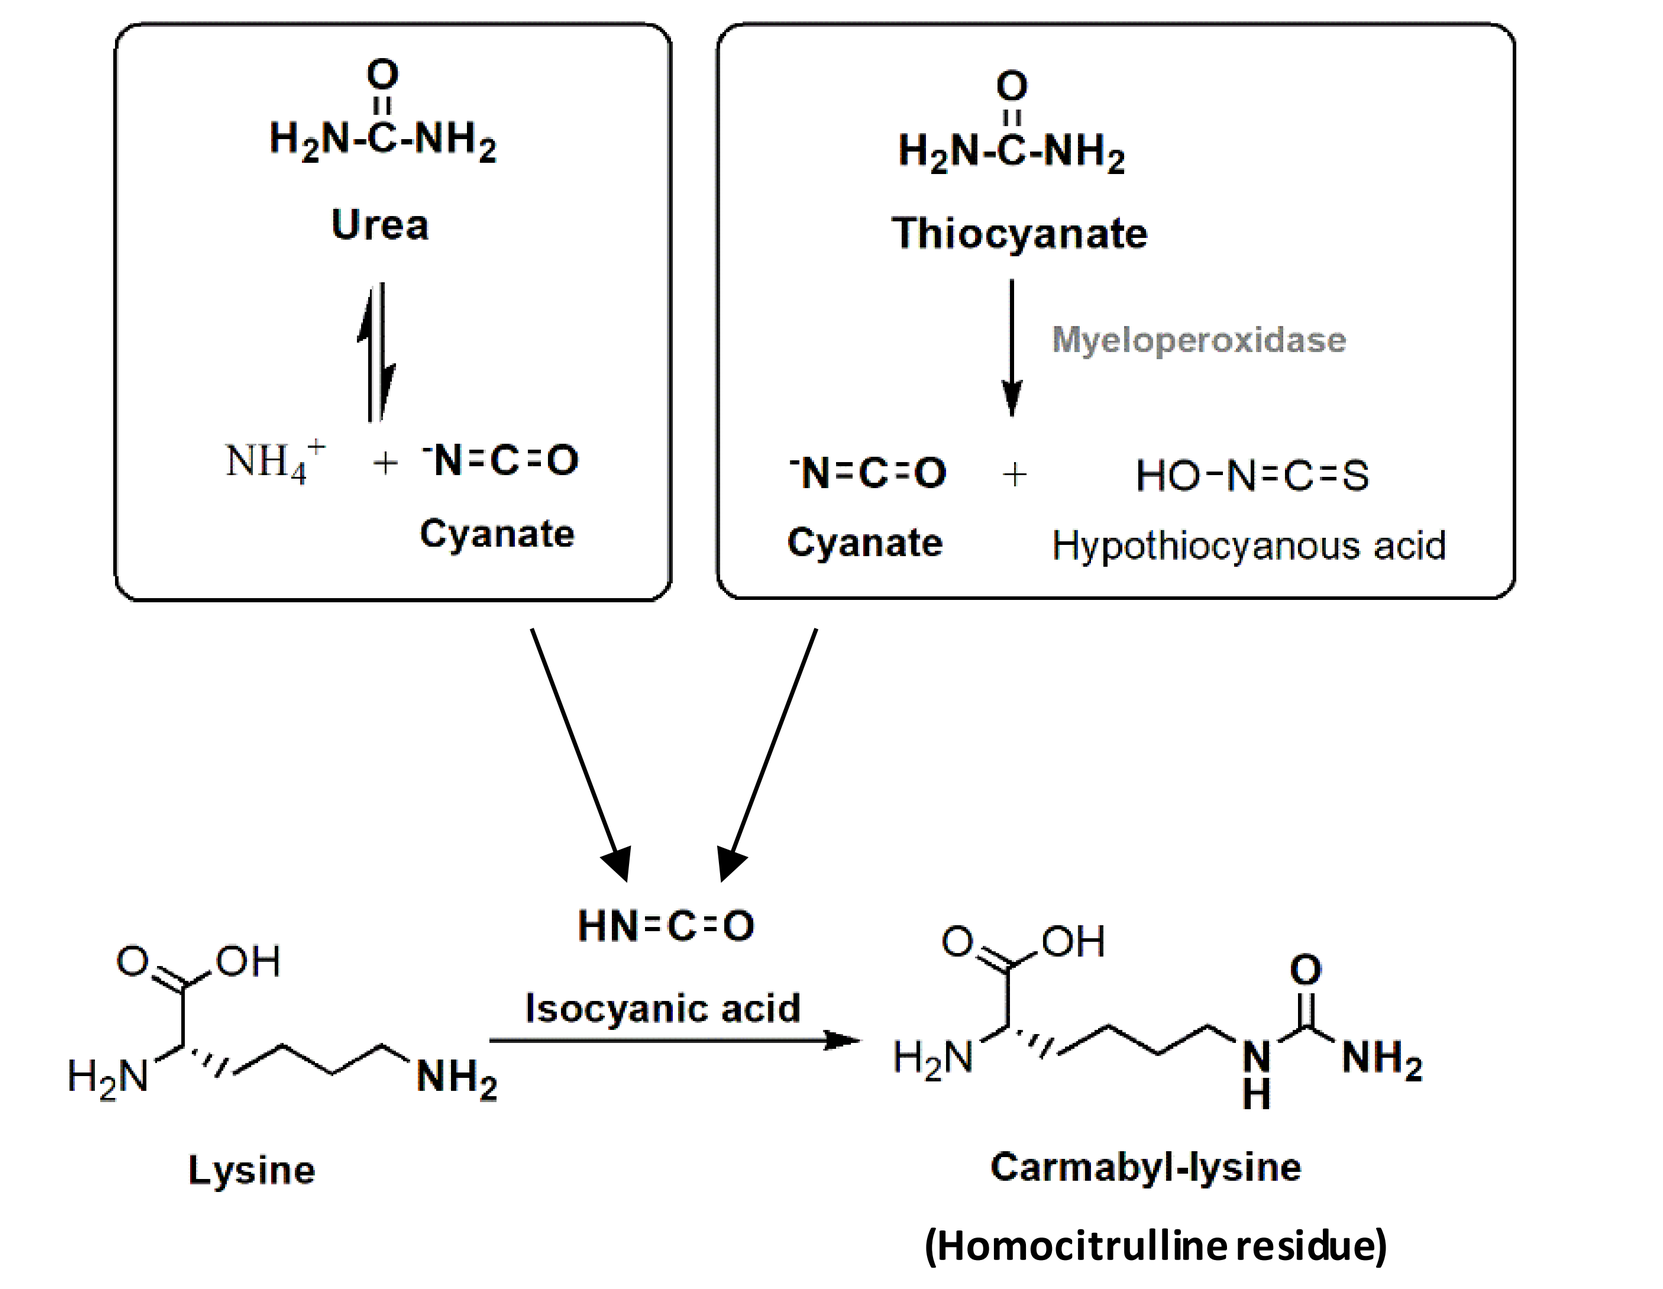
Urea
Thiocyanate
Myeloperoxidase
Cyanate
Hypothiocyanous acid
Isocyanic acid
Lysine
Carmabyl-lysine
(Homocitrulline residue)

Carbamylation. A modification that is driven by increased isocyanic acid levels. This occurs physiologically due to the breakdown of Urea (A) or the actions of MPO enzyme on thiocyanate (B). Carbamylation converts positively charged lysine to neutrally charged homocitrulline.[4]

- Cellular stresses (e.g. proliferating cancer cells) induce autophagy which results in post-translational modifications; one of which is citrullination (cit).
- Citrullination is a post-translational modification of arginine to citrulline by peptidyl deiminase (PAD) enzymes in the presence of high levels of calcium.
- Carbamylation of lysine by isocyanic acid leads to homocitrullination (hcit).
- Myeloperoxidase produced by myeloid suppressor cells catalyzes the build up of isocyanic acid which diffuses into the tumor cells and homocitrullinates cytoplasmic proteins.
- Modified proteins are then degraded and presented by MHC-II for recognition by CD4 T cells.
- In murine models, immunization with modified peptides results in a strong anti-tumour immunity and long-term survival [1-4]
- Here we examine the T cell repertoire to modified peptides in healthy donors and cancer patients.

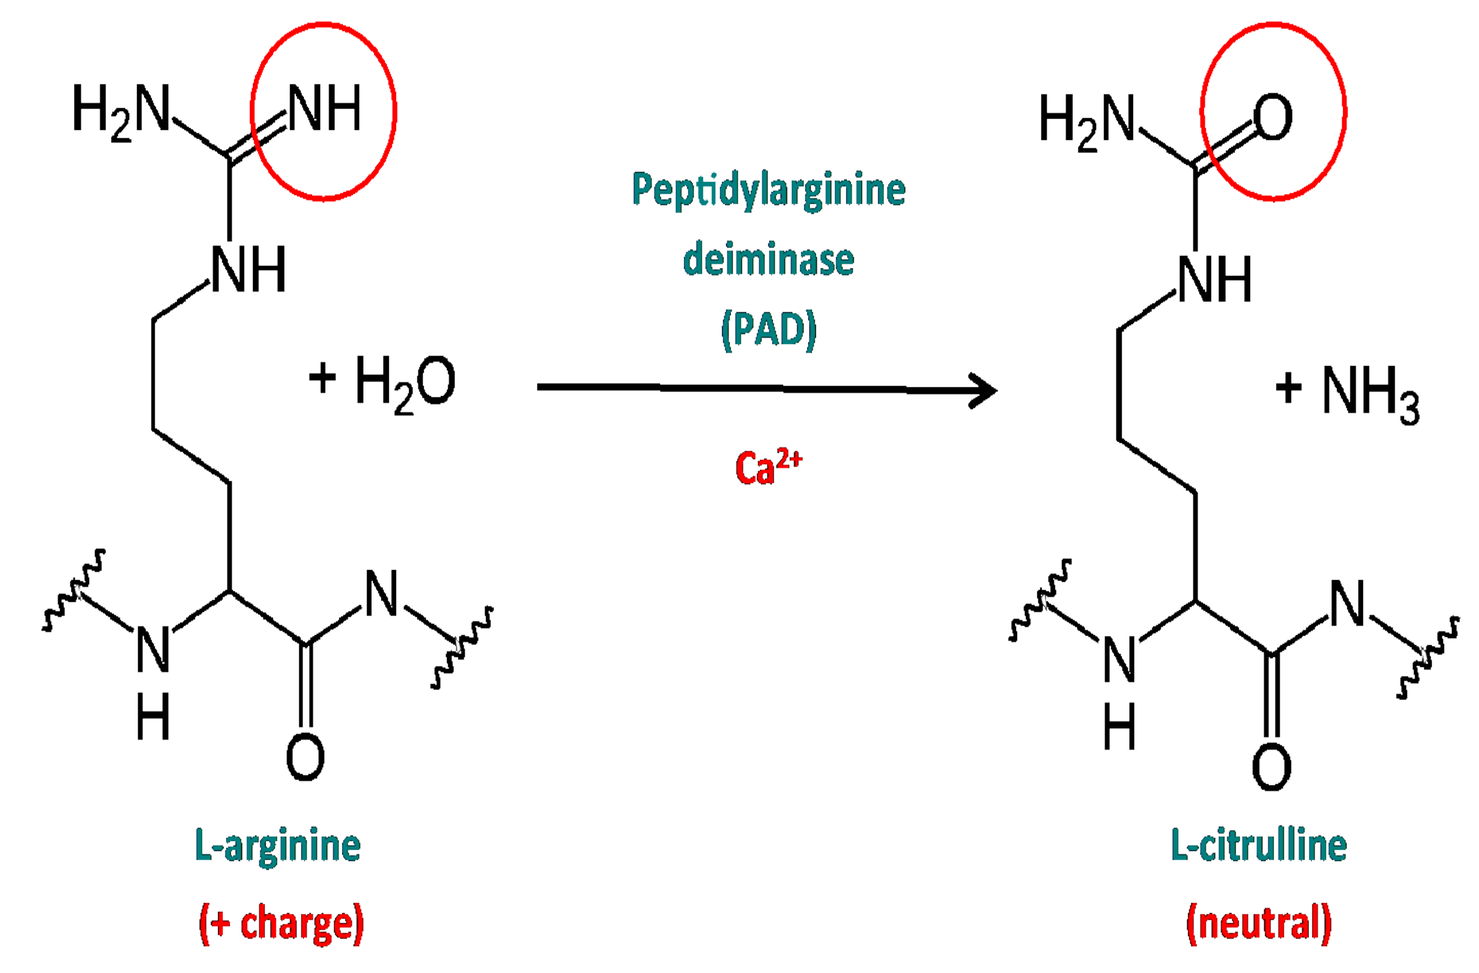
Peptidylarginine deiminase (PAD)
Ca^{2+}
L-arginine
(+ charge)
L-citrulline
(neutral)

Citrullination. A modification that occurs within stressed cells. Peptidylarginine deiminase (PADs) enzymes are activated and convert arginine to citrulline by altering the positively charge aldimine group (=NH) group of arginine to the neutrally charged ketone group (=O) of citrulline.

Healthy donors show a repertoire of CD4 T cells to citrullinated and homocitrullinated peptides which maybe attenuated in cancer patients

A

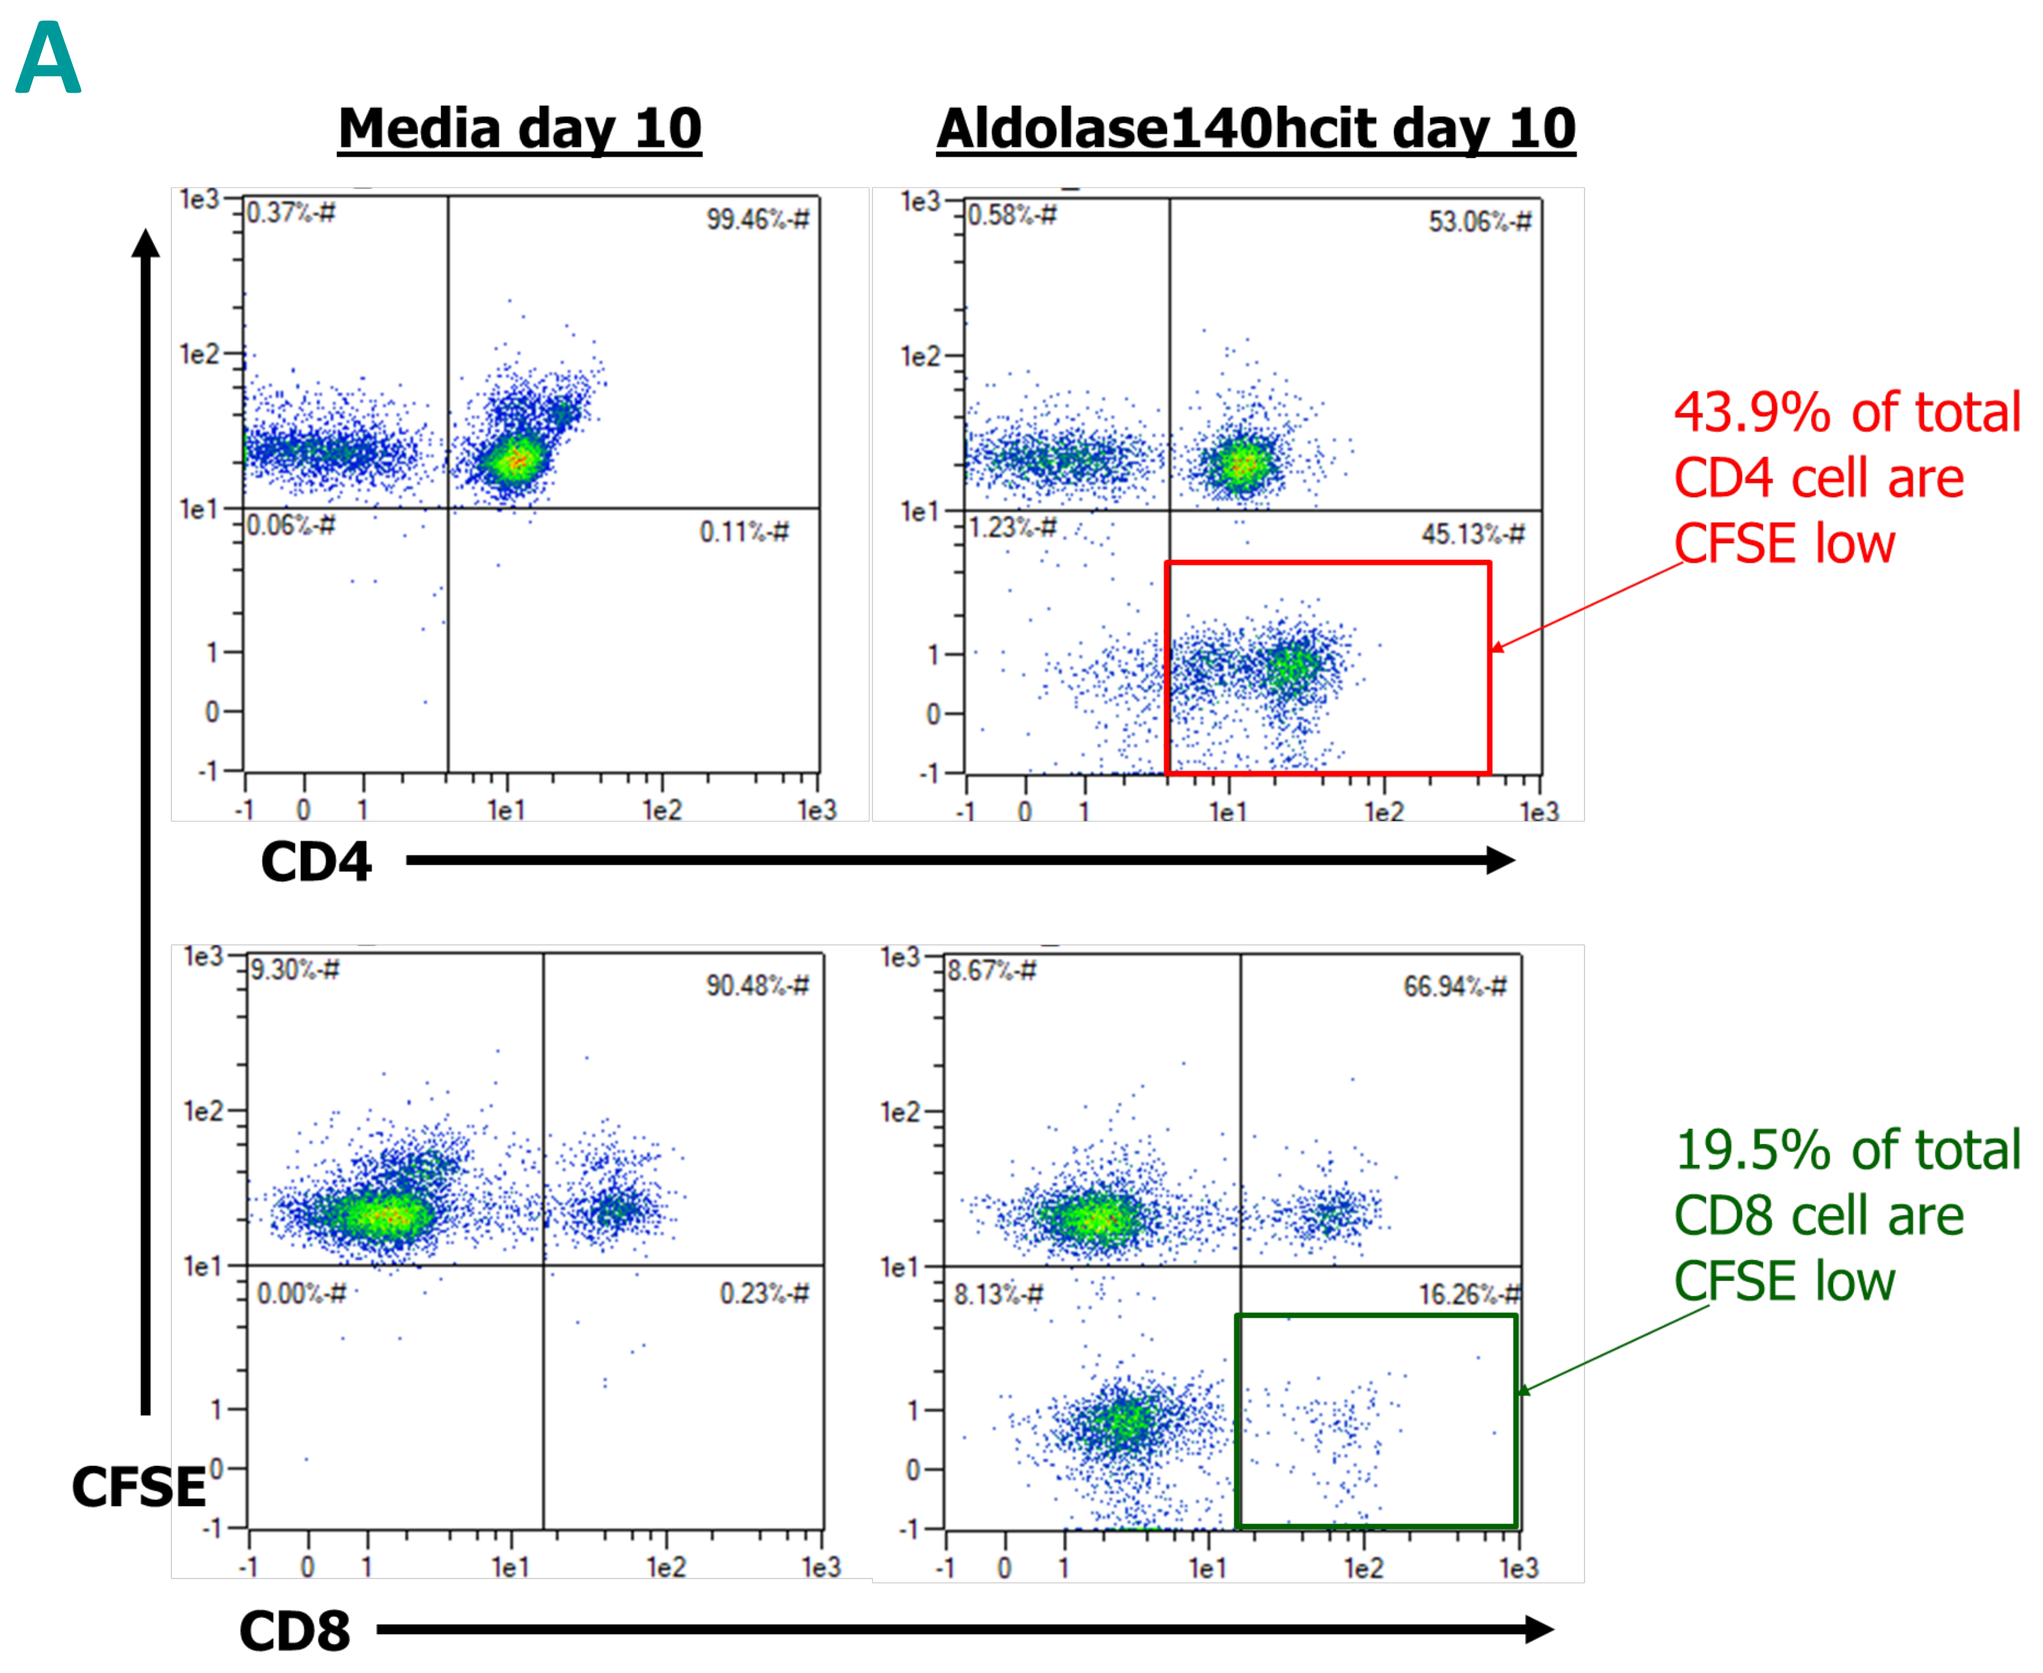
Media day 10
Aldolase140hcit day 10
43.9% of total CD4 cell are CFSE low
19.5% of total CD8 cell are CFSE low
CD4
CD8
CFSE

B

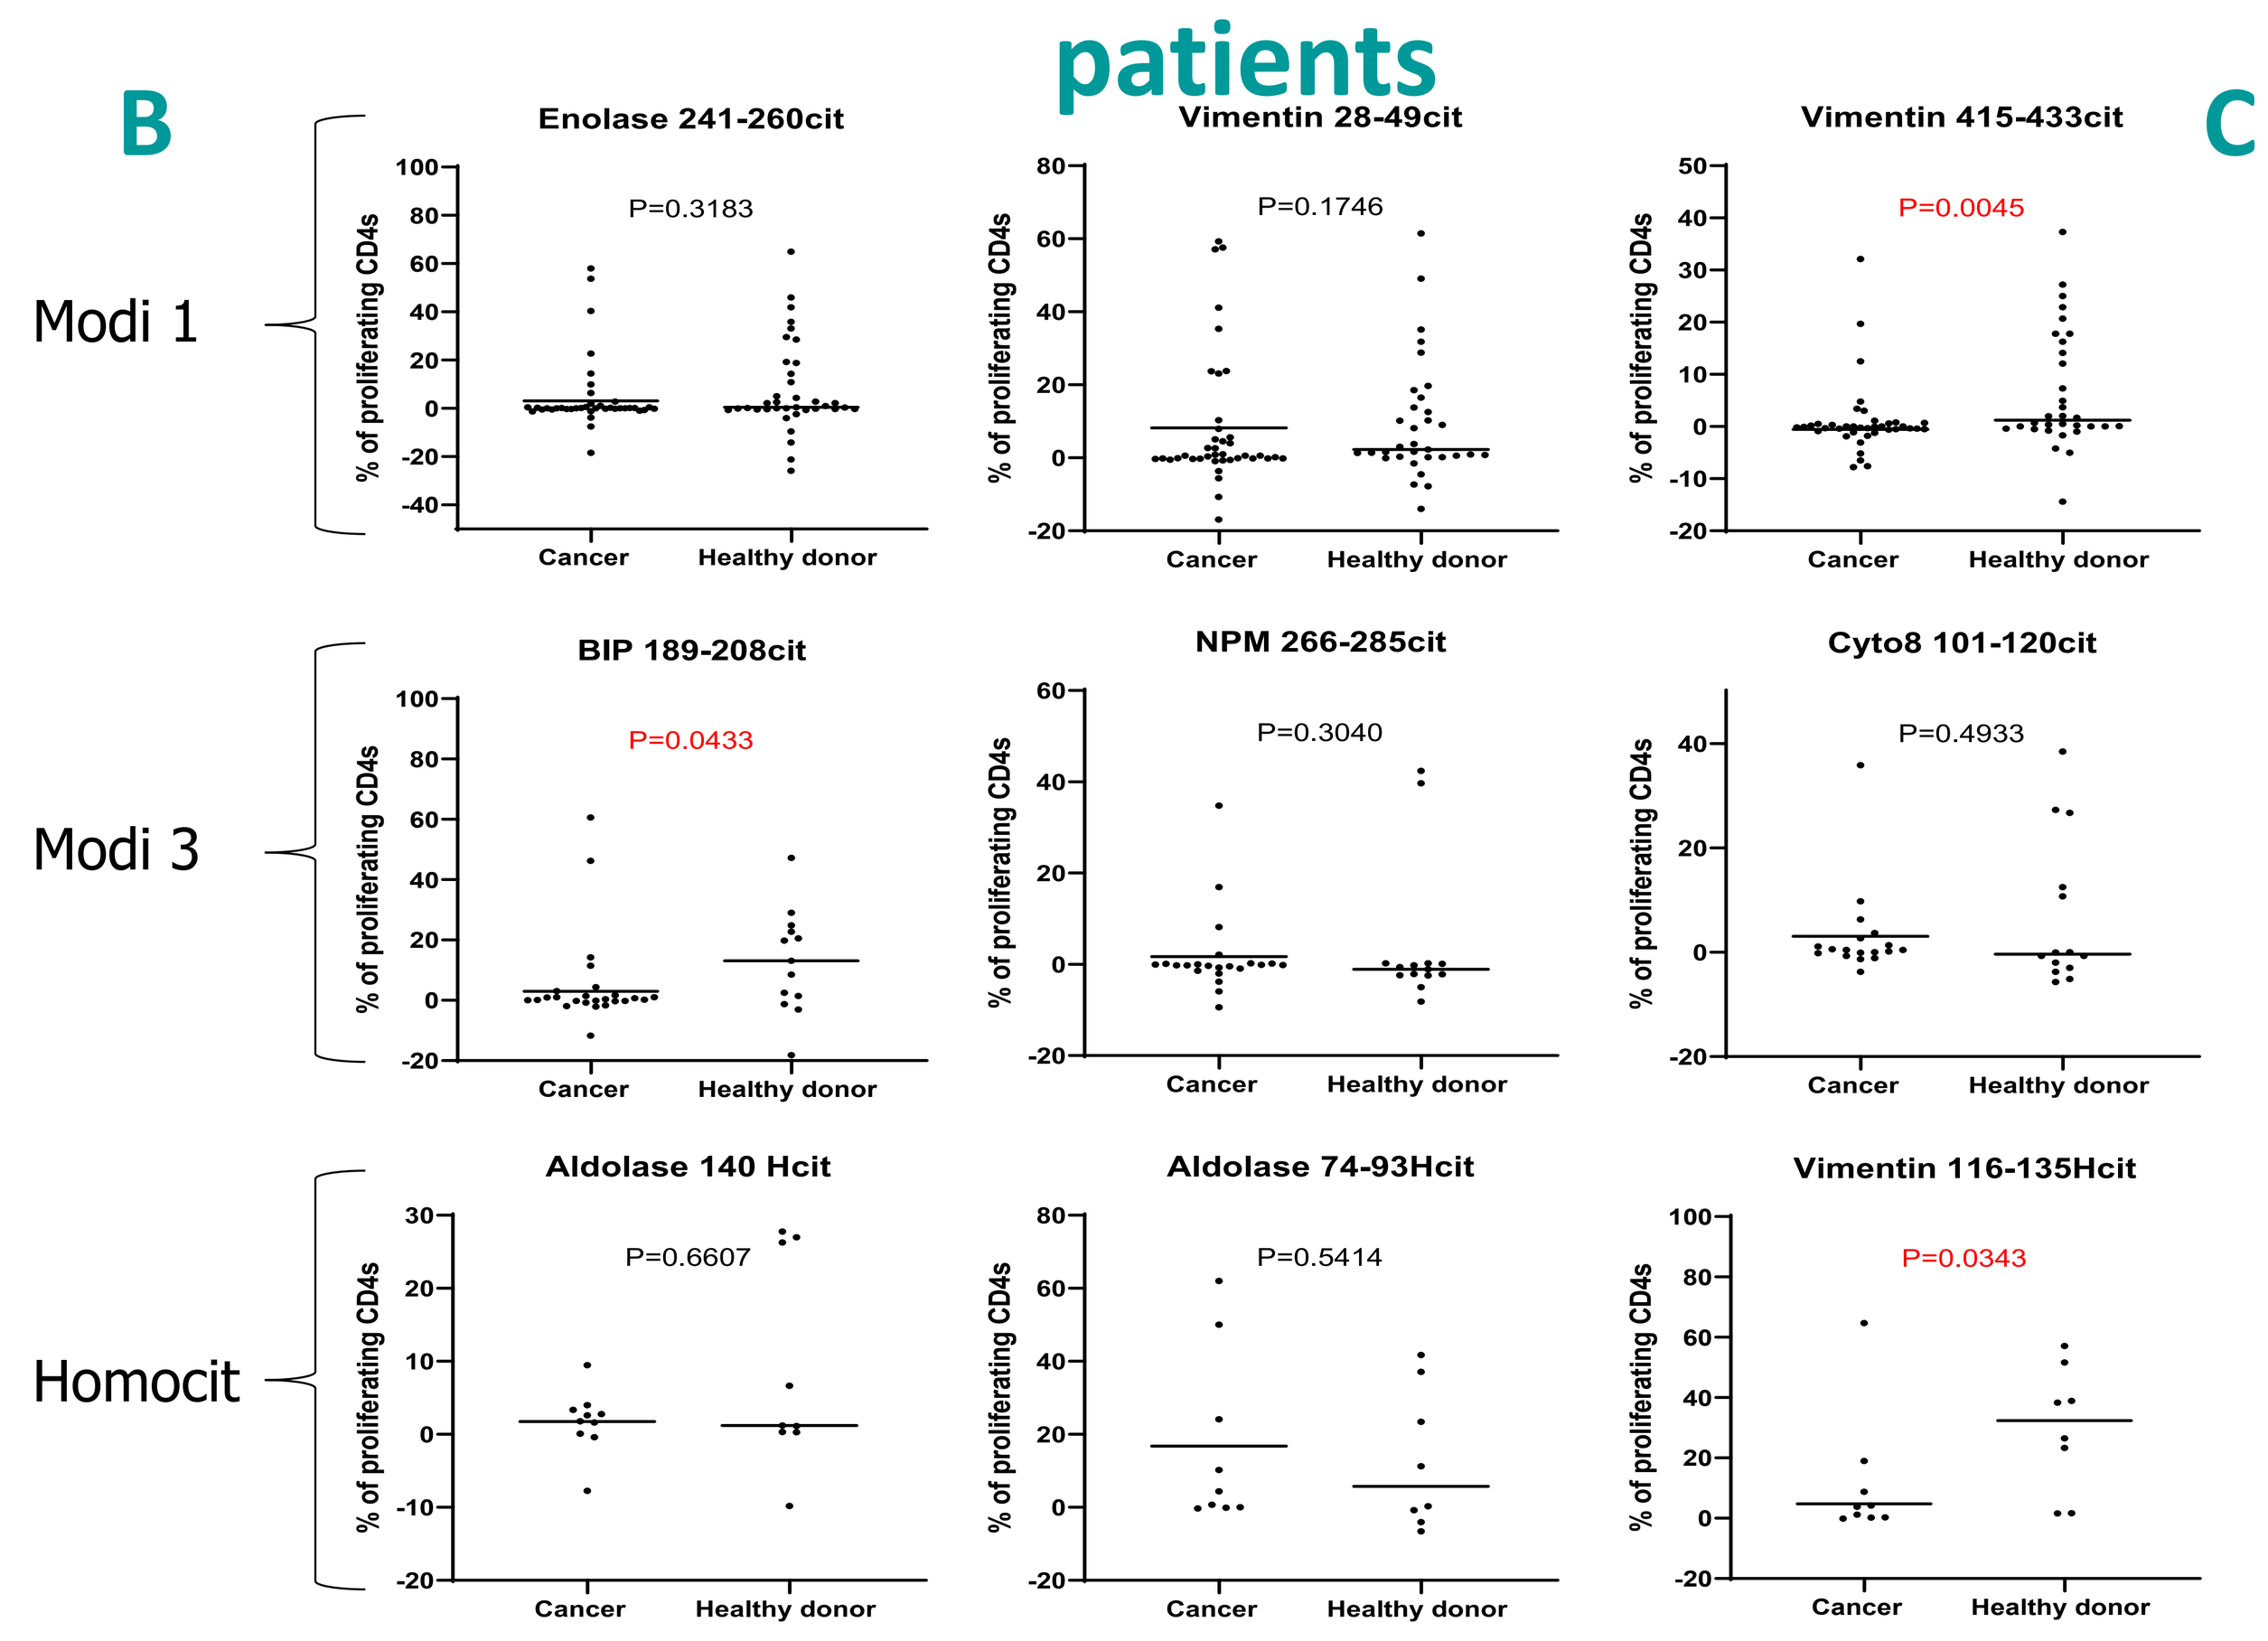
Modi 1
Modi 3
Homocit
Enolase 241-260cit
P=0.3183
Vimentin 28-49cit
P=0.1746
Vimentin 415-433cit
P=0.0045
BIP 189-208cit
P=0.0433
NPM 266-285cit
P=0.3040
Cyto8 101-120cit
P=0.4933
Aldolase 140 Hcit
P=0.6607
Aldolase 74-93Hcit
P=0.5414
Vimentin 116-135Hcit
P=0.0343
% of proliferating CD4s
Cancer
Healthy donor

C

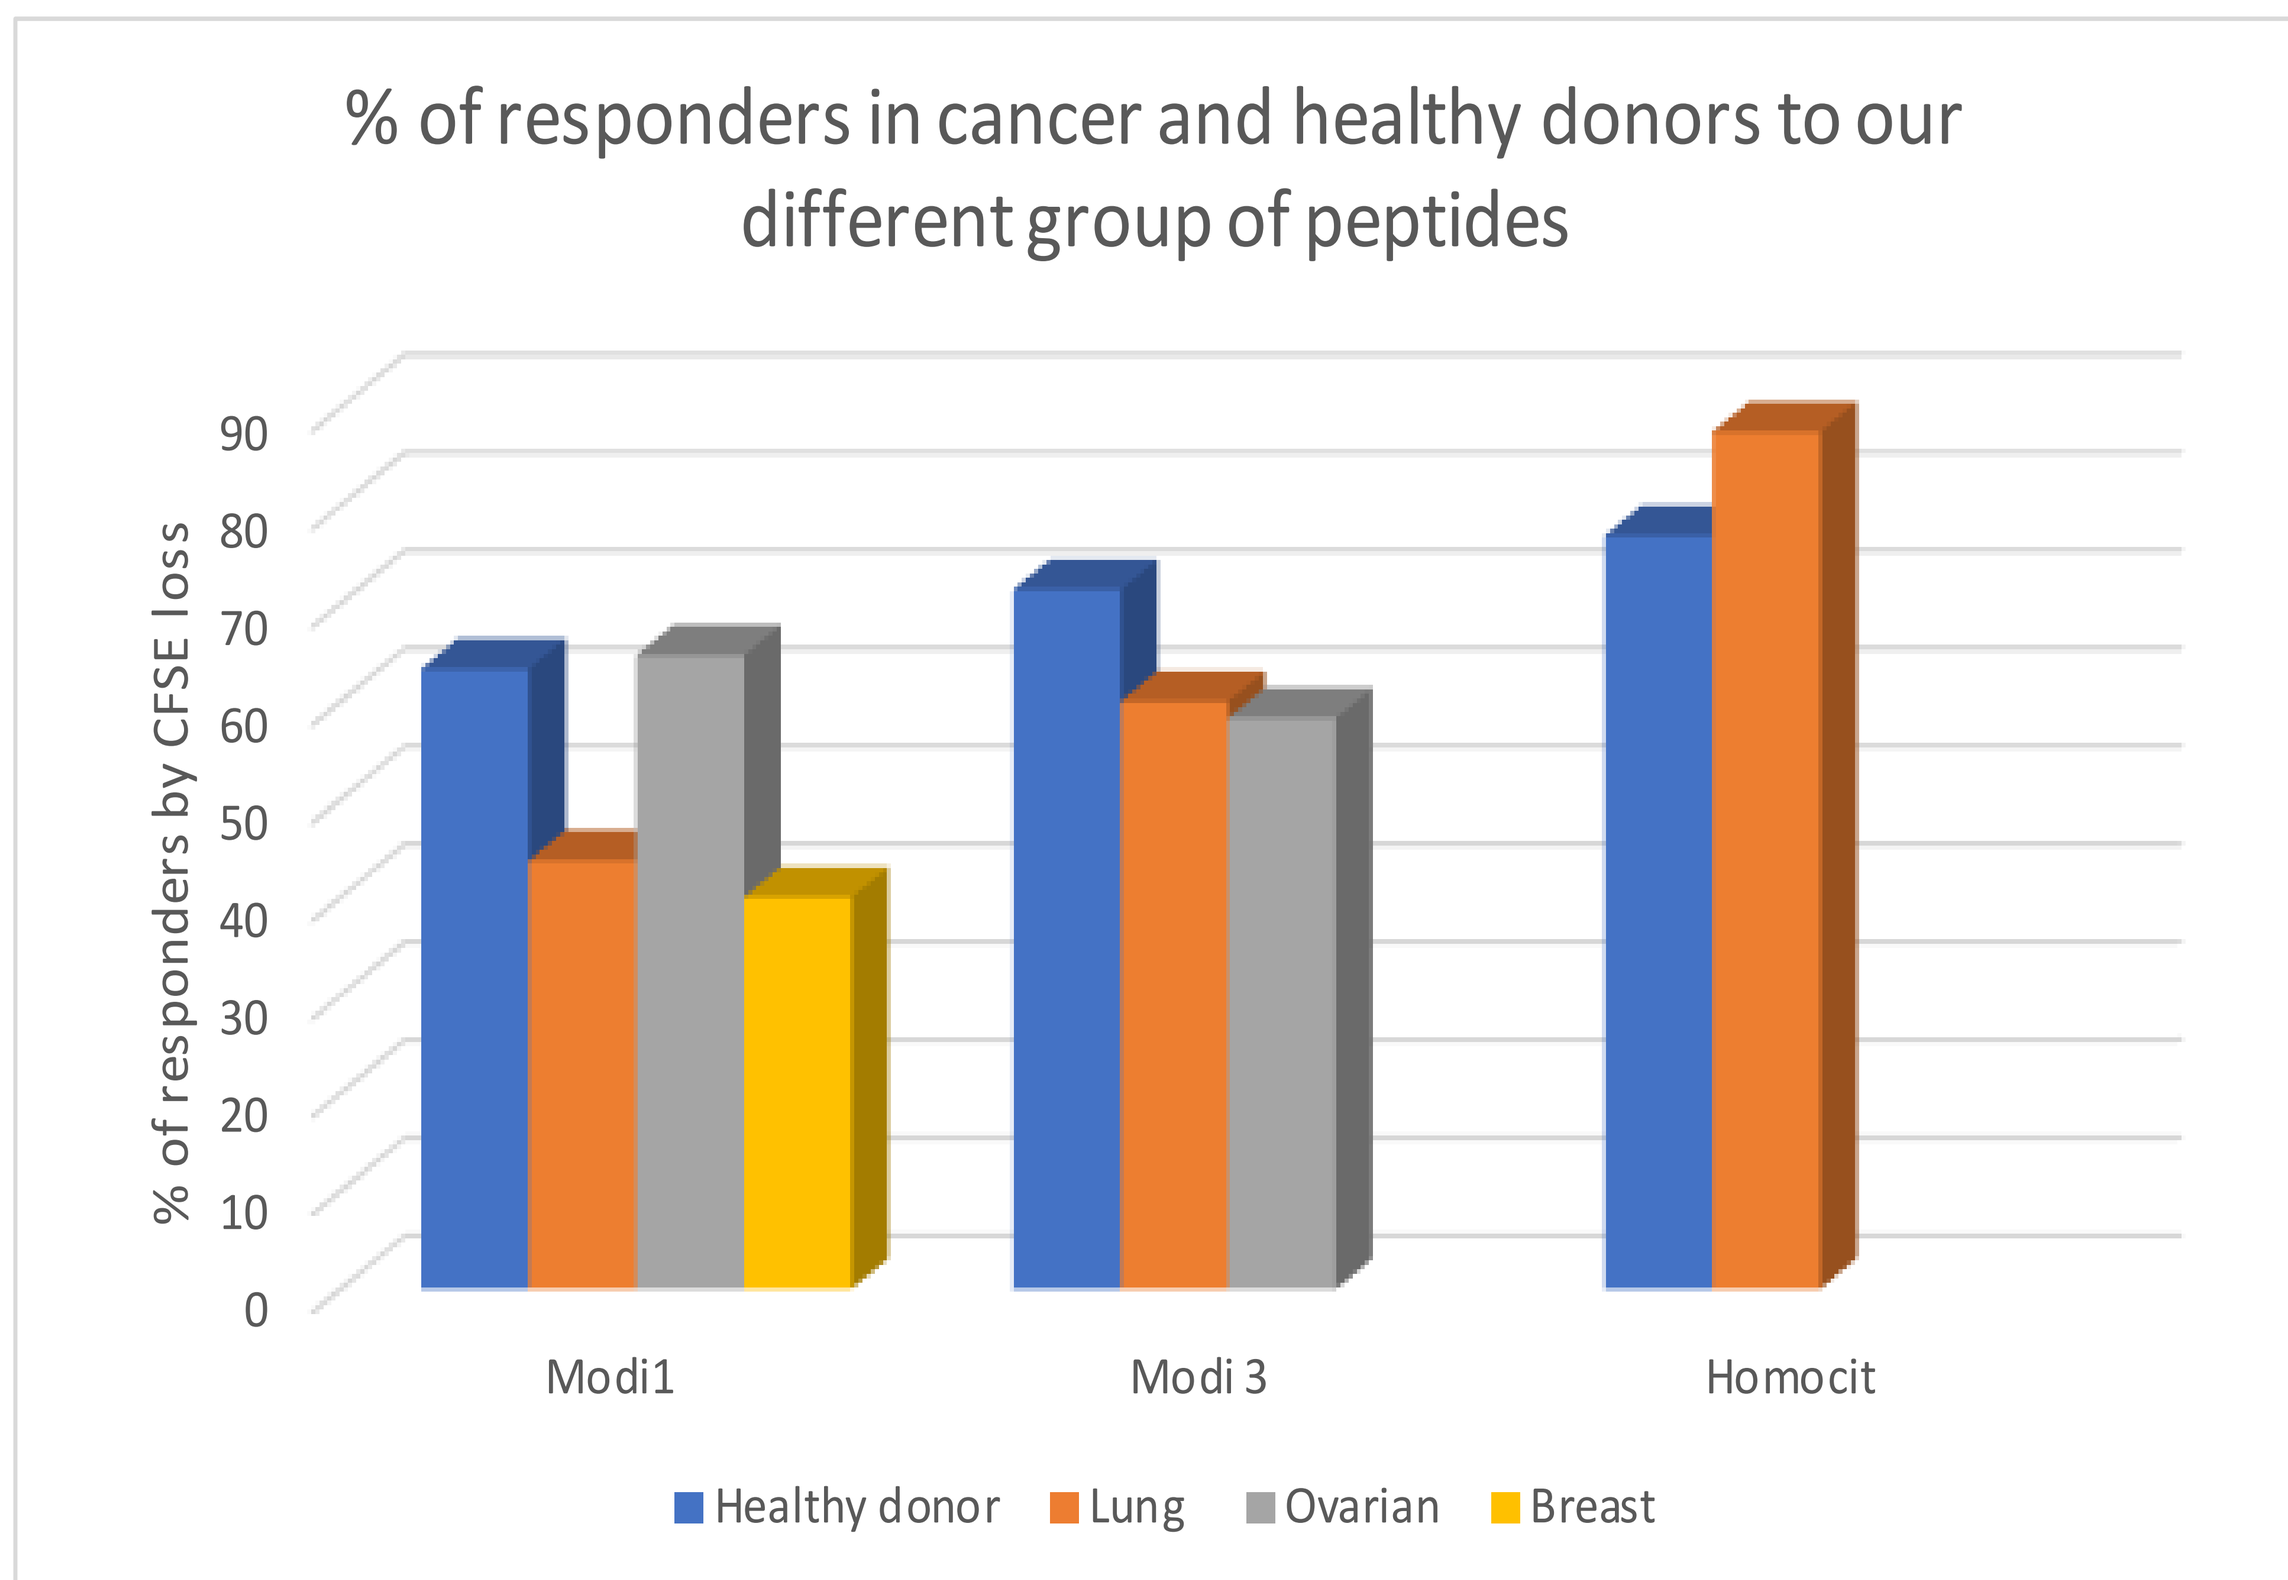
% of responders in cancer and healthy donors to our different group of peptides
% of responders by CFSE loss
Modi1
Modi 3
Homocit
Healthy donor
Lung
Ovarian
Breast

Figure 1: Cancer patient PBMCs tested for responses to cit (modi1 and modi3) and hcit (homocit) peptides by CFSE proliferation assay. A, an example of staining profile for a cancer patient showing specific CD4 cell proliferation (CFSE loss) to homocitrullinated aldolase-140. Response is defined as double the negative background control and above 1%. B, comparison of the percentage of proliferating CD4 cells in a number of healthy donors and cancer patients in response to the different citrullinated and homocitrullinated peptides. All data was normalised against background negative control. C, percentage of responding donors to at least one peptide (healthy donors vs lung cancer vs breast cancer vs ovarian cancer).

Activation marker CD71 can be used to define responses but higher LAP expression in cancer patients maybe attenuating proliferative responses

Figure 2. Example showing relation between CD71⁺ and CFSE^low^ staining in cancer compared to healthy donors (gated on CD4+ cells) and phenotyping of responding CD4+CD71+ population for expression of LAP

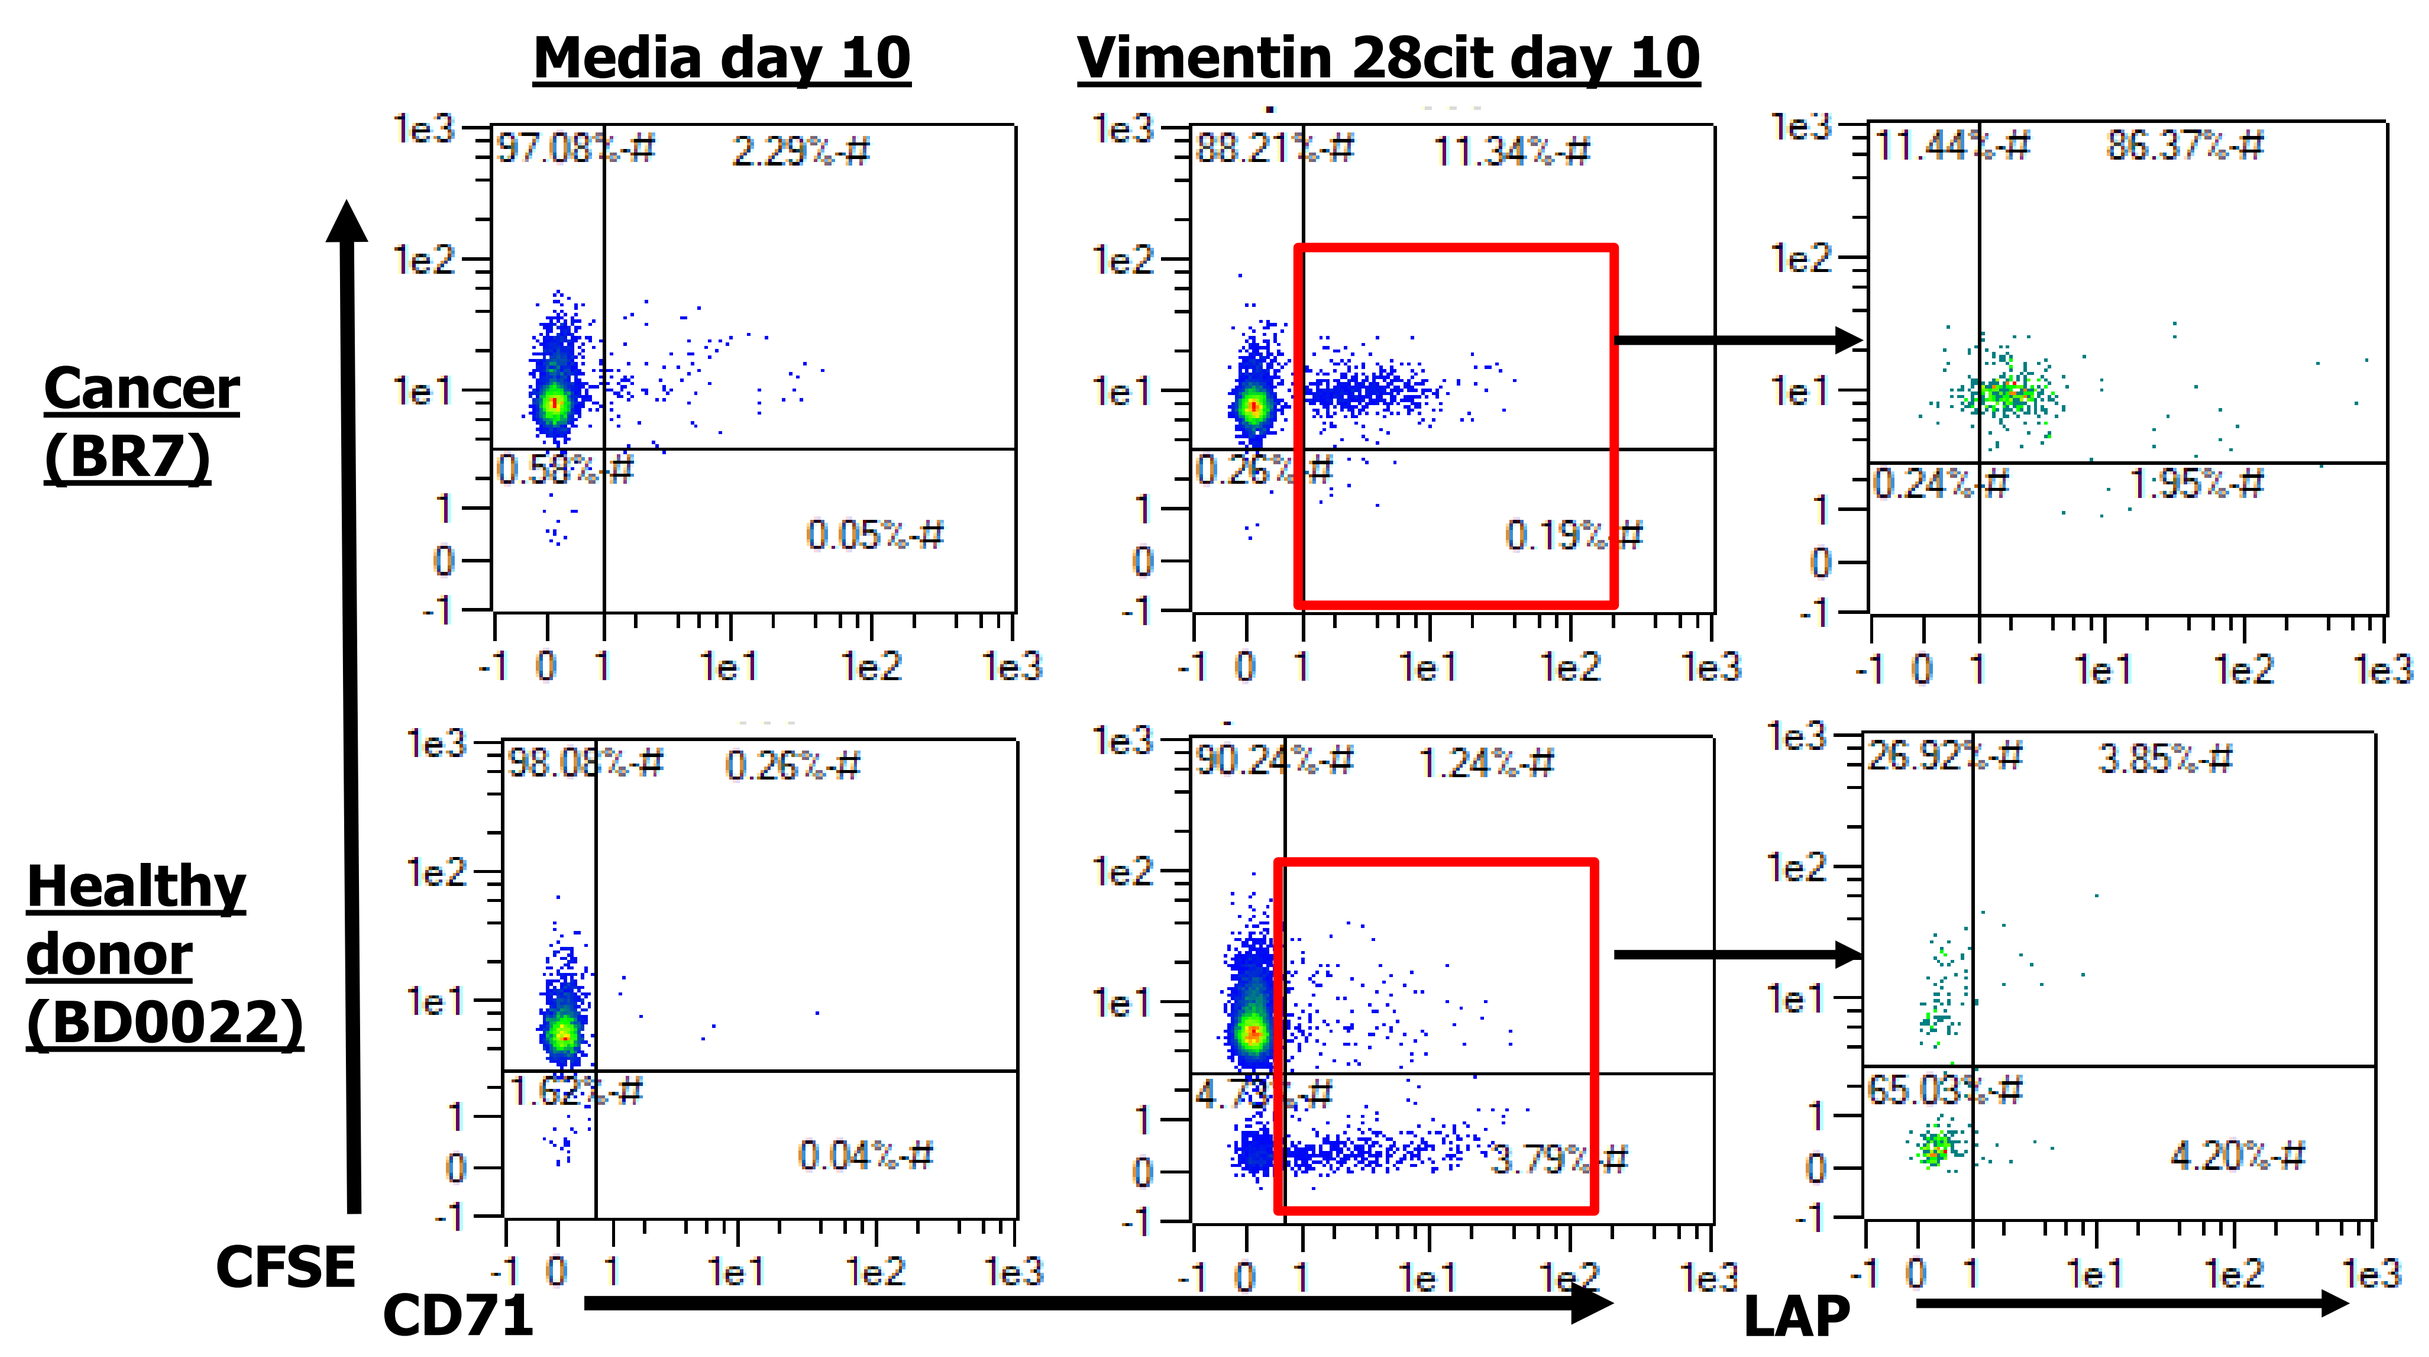
Media day 10
Vimentin 28cit day 10
Cancer (BR7)
Healthy donor (BD0022)
CFSE
CD71
LAP

- Latency associated peptide (LAP) and transforming growth factor- β (TGF-β) are present as an intracellular homodimeric complex. Cleavage occurs by proteases but they remain non-covalently associated. This is called the small latent complex (SLC)
- SLC then covalently binds with latent TGF-β binding protein (LTβP) to form the large latent complex (LLC) which is expressed in the extracellular membrane.
- Only some cells activate TGF-β through integrins, thrombospondin-1 and proteases.
- Cancer patients are have a regulatory $CD4^+LAP^+Foxp3^-$ population both in the PBMCs and tumur microenvironment which is more suppressive than conventional T-regs[5].
- Here we show an antigen specific population which maybe attenuating responses.

Cancer patients have a higher population of LAP⁺ antigen specific T cells and the expression of LAP is inversely correlated with proliferative responses

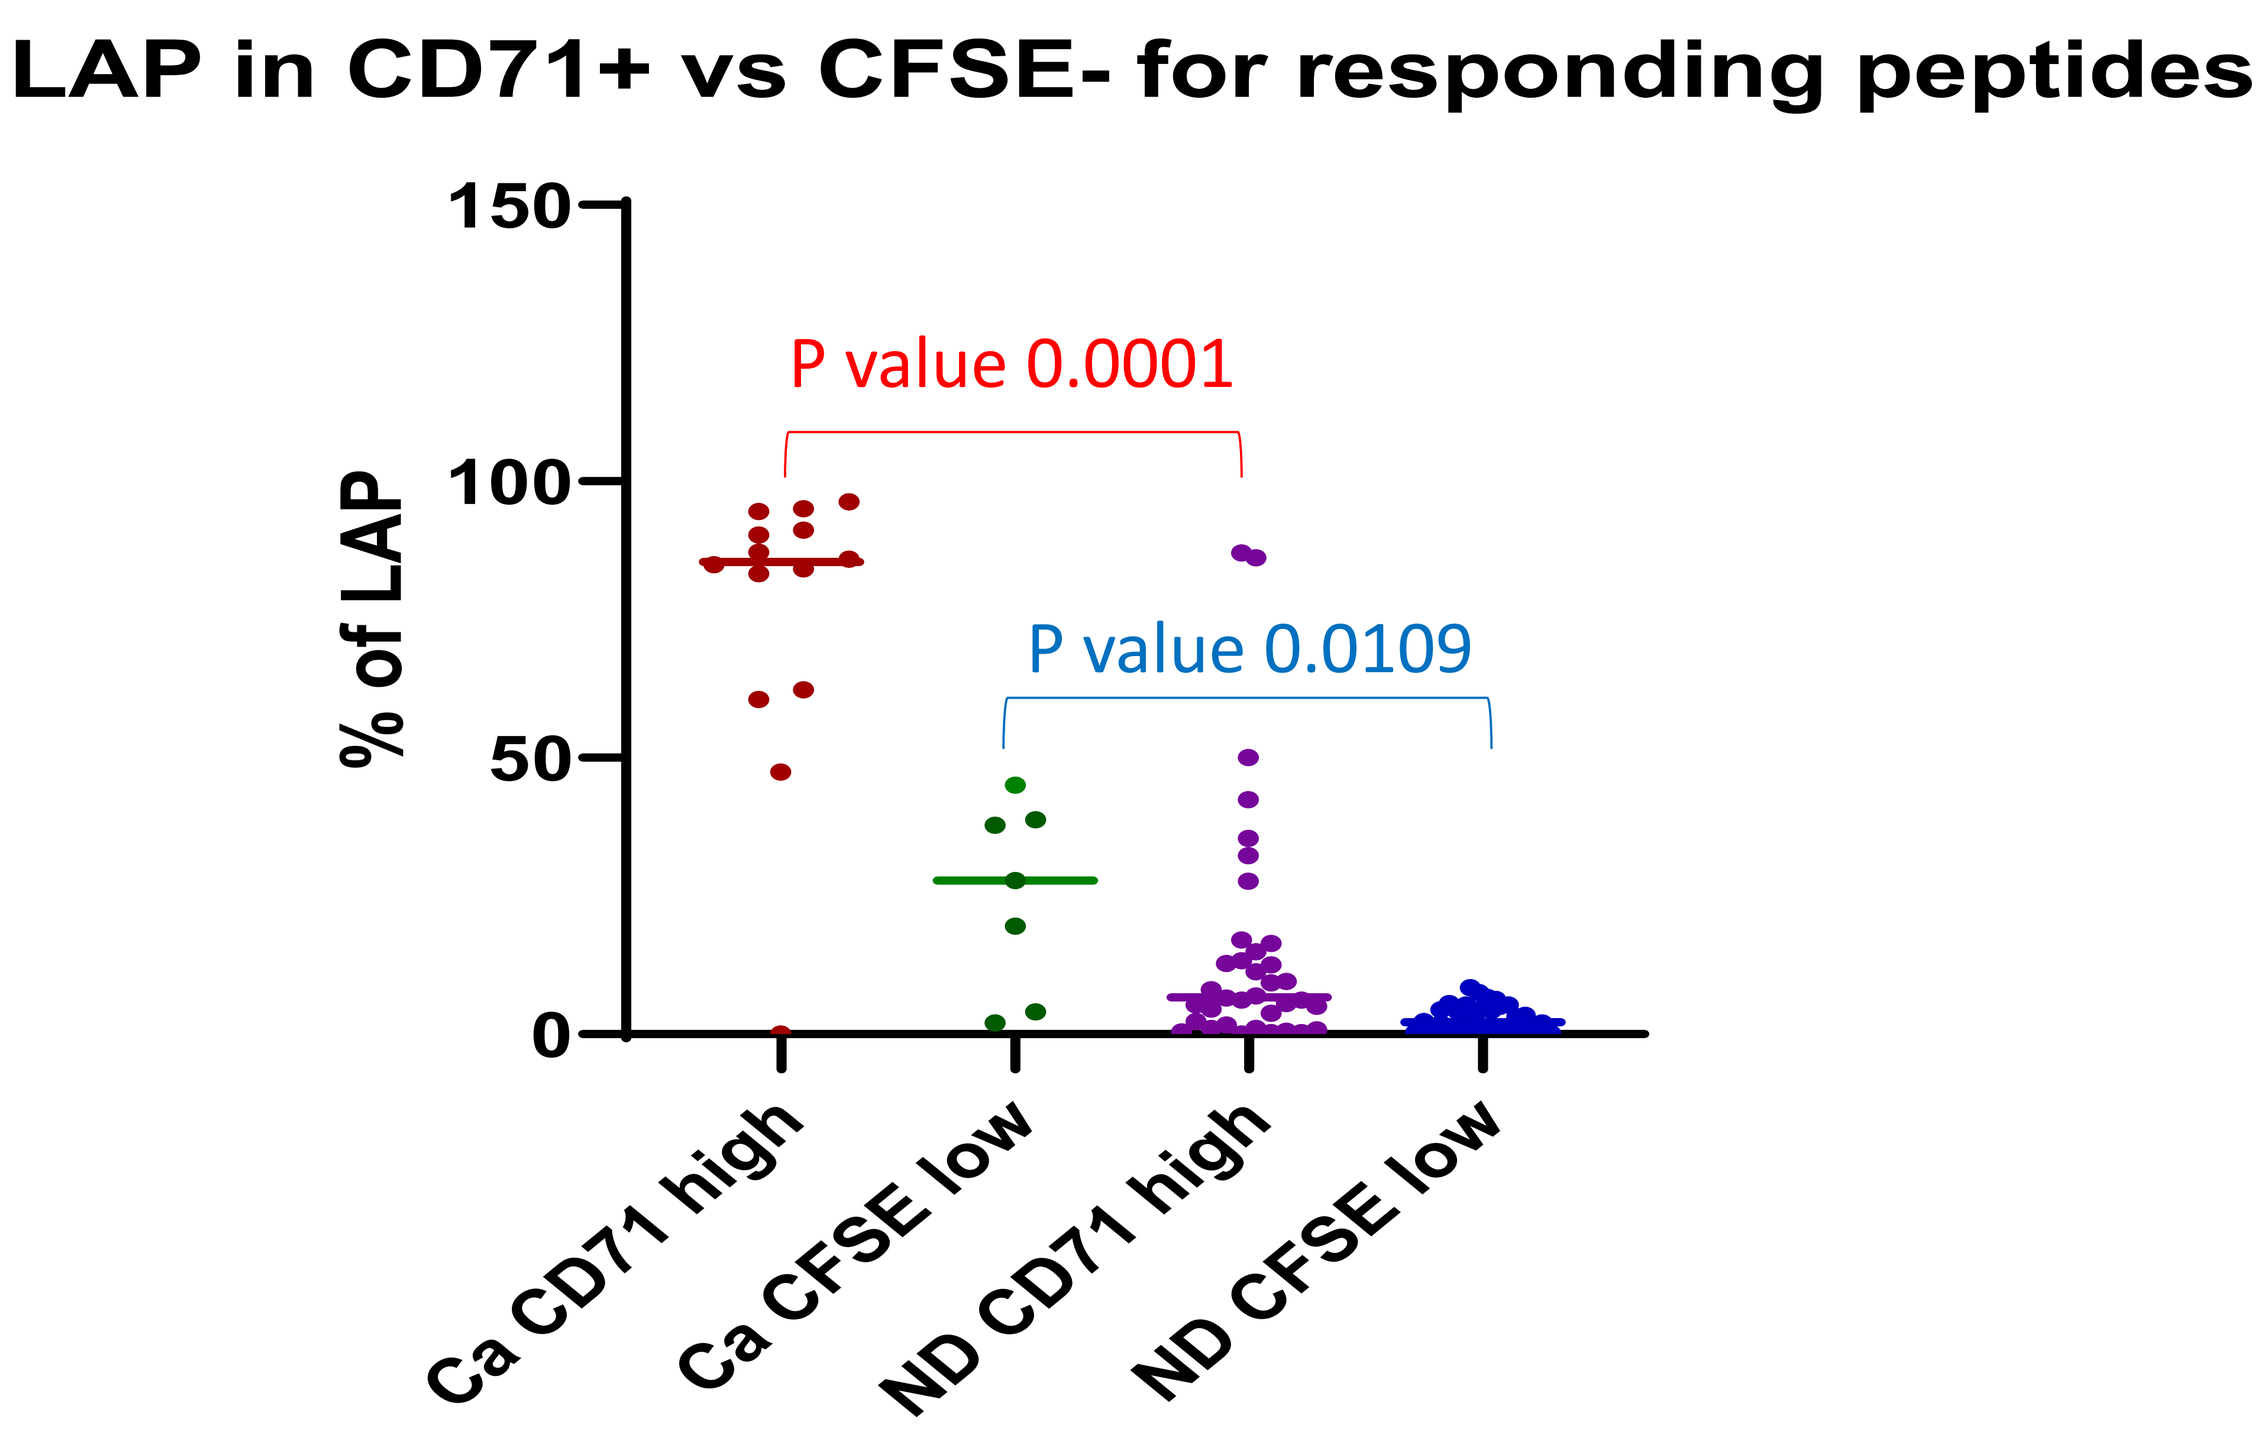
LAP in CD71+ vs CFSE- for responding peptides
% of LAP
P value 0.0001
P value 0.0109
Ca CD71 high
Ca CFSE low
ND CD71 high
ND CFSE low

Figure 3. LAP expression on responding populations ($CD4^+CD71^+$ or $CD4^+CFSE^{low}$) is higher in cancer patients (Ca) compared to healthy donors (ND).

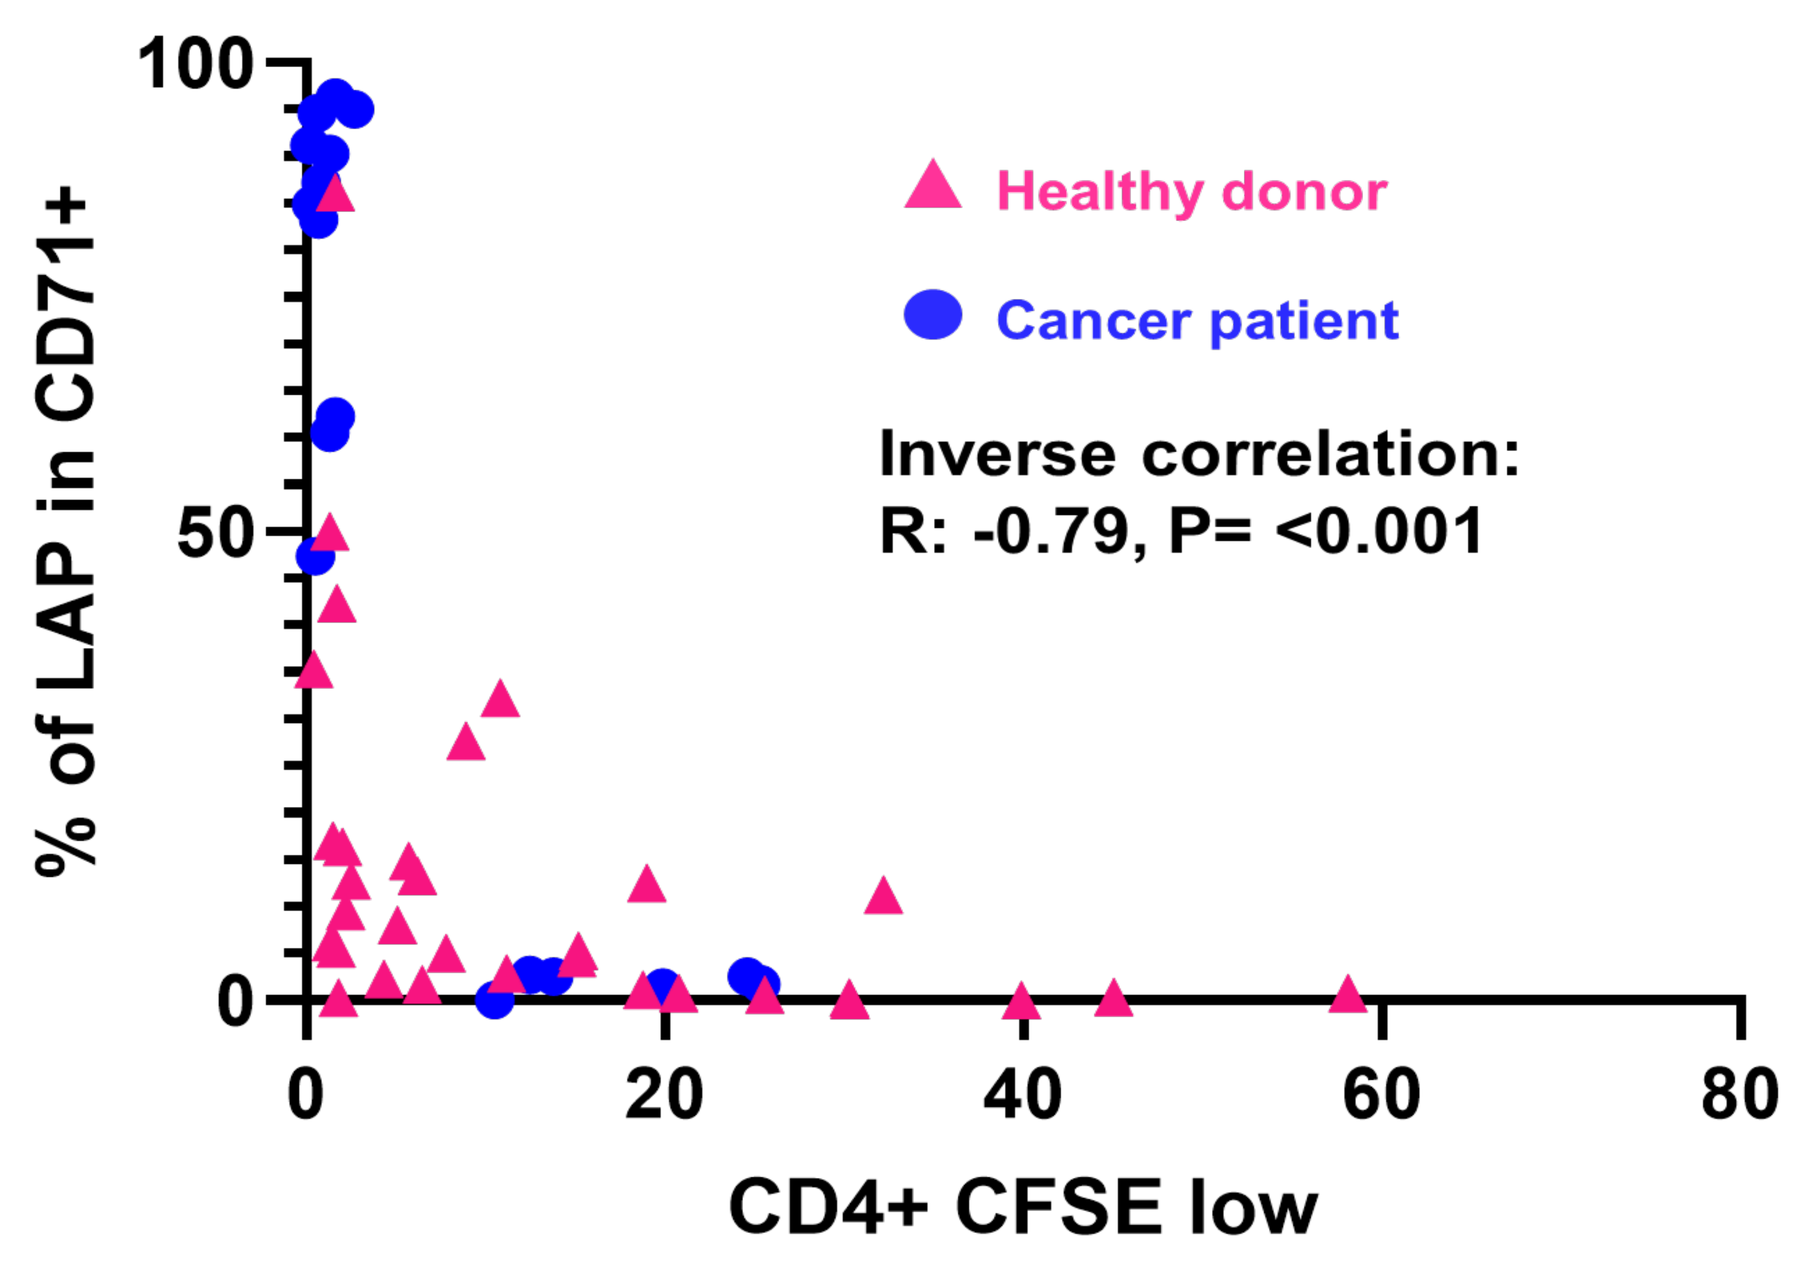
% of LAP in CD71+
CD4+ CFSE low
Healthy donor
Cancer patient
Inverse correlation:
R: -0.79, P= <0.001

Figure 4. Graph showing inverse correlation between proliferation ($CD4^+$ $CFSE^{low}$) and expression of LAP in activated $CD4^+CD71^+$ cells, both in healthy donors and cancer patients.

CONCLUSIONS

- Cancer patients' PBMCs show a specific $CD4^+$ repertoire when cultured with citrullinated and homocitrullinated peptides comparable to healthy donors but maybe attenuated.
- CD71 expression can be used to identify activation in response to the peptides which does not always correlate with a proliferative response.
- Antigen specific $CD4^+CD71^+$ cells and $CD4^+CFSE^{low}$ cells have significantly more LAP expression in cancer patients compared to healthy donors.
- $CD4^+CD71^+LAP^+$ population has an inverse correlation with proliferation.
- Modified antigens maybe a good target for anti-tumor immunity but cancer patients have a specific LAP^+ regulatory population which would either need to be inhibited with a LAP/TGF-β neutralizing antibody or polarized to Th1 with vaccination.

References:

1. Brentville, V.A., et al., *Citrullinated Vimentin Presented on MHC-II in Tumor Cells Is a Target for CD4+ T-Cell-Mediated Antitumor Immunity.* Cancer Res, 2016. **76**(3): p. 548-60.
2. Brentville, V.A., et al., *T cell repertoire to citrullinated self-peptides in healthy humans is not confined to the HLA-DR SE alleles; Targeting of citrullinated self-peptides presented by HLA-DP4 for tumour therapy.* Oncoimmunology, 2019. **8**(5): p. e1576490.
3. Cook, K., et al., *Citrullinated alpha-enolase is an effective target for anti-cancer immunity.* Oncoimmunology, 2018. **7**(2): p. e1390642.
4. Durrant, L.G., R.L. Metheringham, and V.A. Brentville, *Autophagy, citrullination and cancer.* Autophagy, 2016. **12**(6): p. 1055-6.
5. Scurr, M., et al., *Highly prevalent colorectal cancer-infiltrating LAP+ Foxp3− T cells exhibit more potent immunosuppressive activity than Foxp3+ regulatory T cells.* Mucosal Immunology, 2013. **7**: p. 428.

PRINTED BY SCIENCEPOSTERS .co.uk