Carbamylation of lysine residues mediated by MDSCs in the tumour environment make excellent targets for CD4 T cell mediated cancer immunotherapy

K Cook[1], W Xue[1], I Daniels[1], P Symonds[1], IM Gijon[1], D. Boocock[3], C. Coveney[3], A. Miles[3], P Vaghela[1], R. Choudhury[1], S Shah[1], S Atabani[1], R Metheringham[1], V Brentville[1], L Durrant[1,2]

[1]Scancell Holdings plc, Oxford, UK, [2]Nottingham University, Nottingham, UK, [3]Nottingham Trent University, UK

INTRODUCTION

- Targeting post translationally modified epitopes may provide new targets for cancer vaccines that avoid the attenuation often seen with responses to self antigens.[1, 2]
- Carbamylation of lysine residues leads to formation of homocitrulline (Hcit), converting the positively charged amino acid to a neutral amino acid which can lead to generation of a new epitope that is recognised by T-cells and B-cells.
- Carbamylation can be driven by myeloperoxidase (MPO) an enzyme produced by neutrophils, macrophages and myeloid derived suppressor cells (MDSCs).[3]
- Two proteins that can be carbamylated are the cytoskeletal protein vimentin and the glycolytic protein aldolase, both of which can be upregulated in cancer cells.
- In this study we demonstrate that Hcit specific CD4 responses can be generated and that these have an anti-tumour effect which is dependent on MDSC populations and MHC II presentation on the tumour.

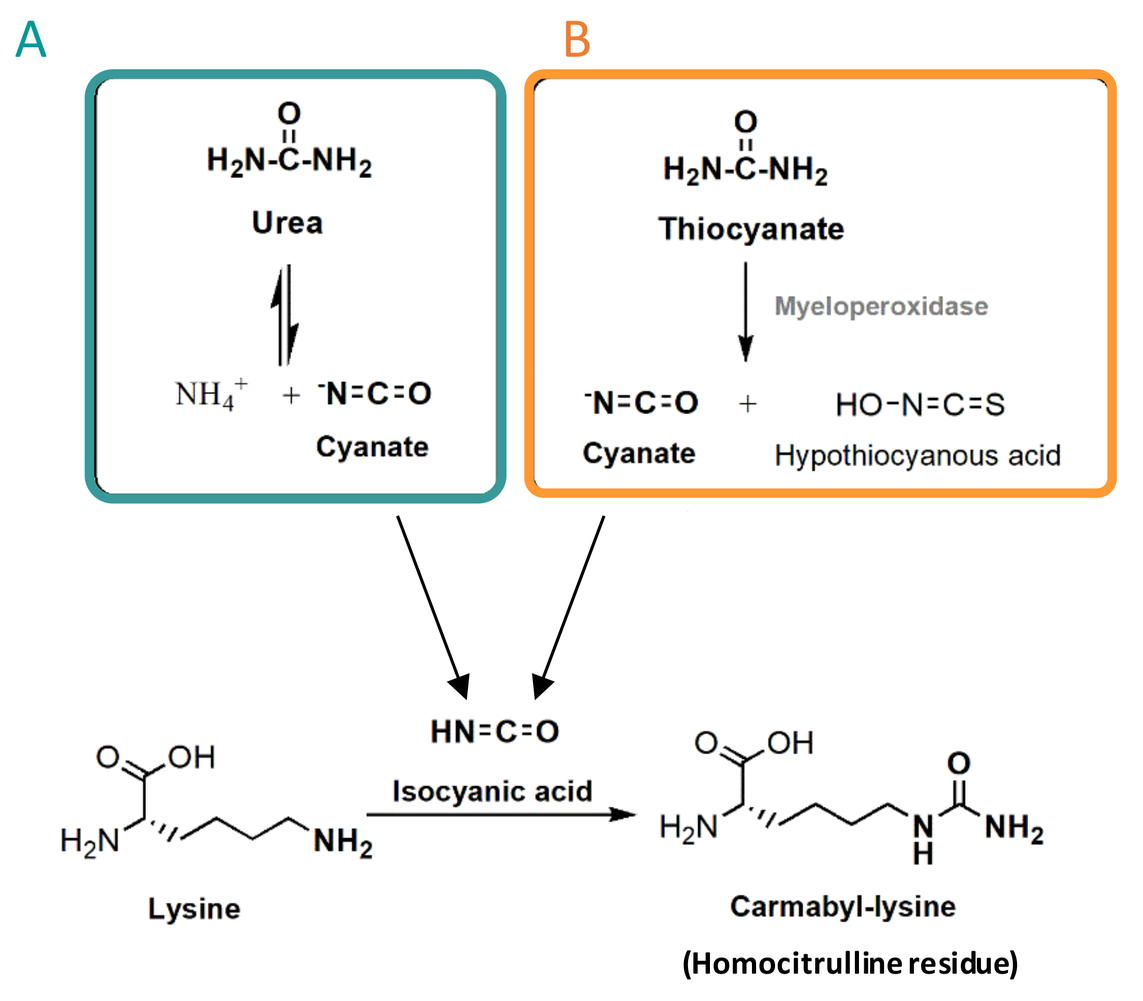
A
B
Urea
Cyanate
Thiocyanate
Myeloperoxidase
Hypothiocyanous acid
Isocyanic acid
Lysine
Carmabyl-lysine
(Homocitrulline residue)

Carbamylation. A modification that is driven by increased isocyanic acid levels. This occurs physiologically due to the breakdown of Urea (A) or the actions of MPO enzyme on thiocyanate (B). Carbamylation converts positively charged lysine to neutrally charged homocitrulline.[4]

Homocitrullinated peptides stimulate strong CD4 responses *in vivo*

- Studies were carried out using transgenic mice expressing different human HLA types.
- Mice were immunised with Hcit peptides with adjuvant CpG/MPLA.
- IFNγ responses were then detected by ELISpot.
- HLA-specific responses were observed to the three peptides used.

	HHDII/DP4	HHDII/DR1	DR4
Aldolase 74Hcit	✓	✗	✗
Aldolase 140Hcit	✓	✓	✗
Vimentin 116Hcit	✗	✓	✓

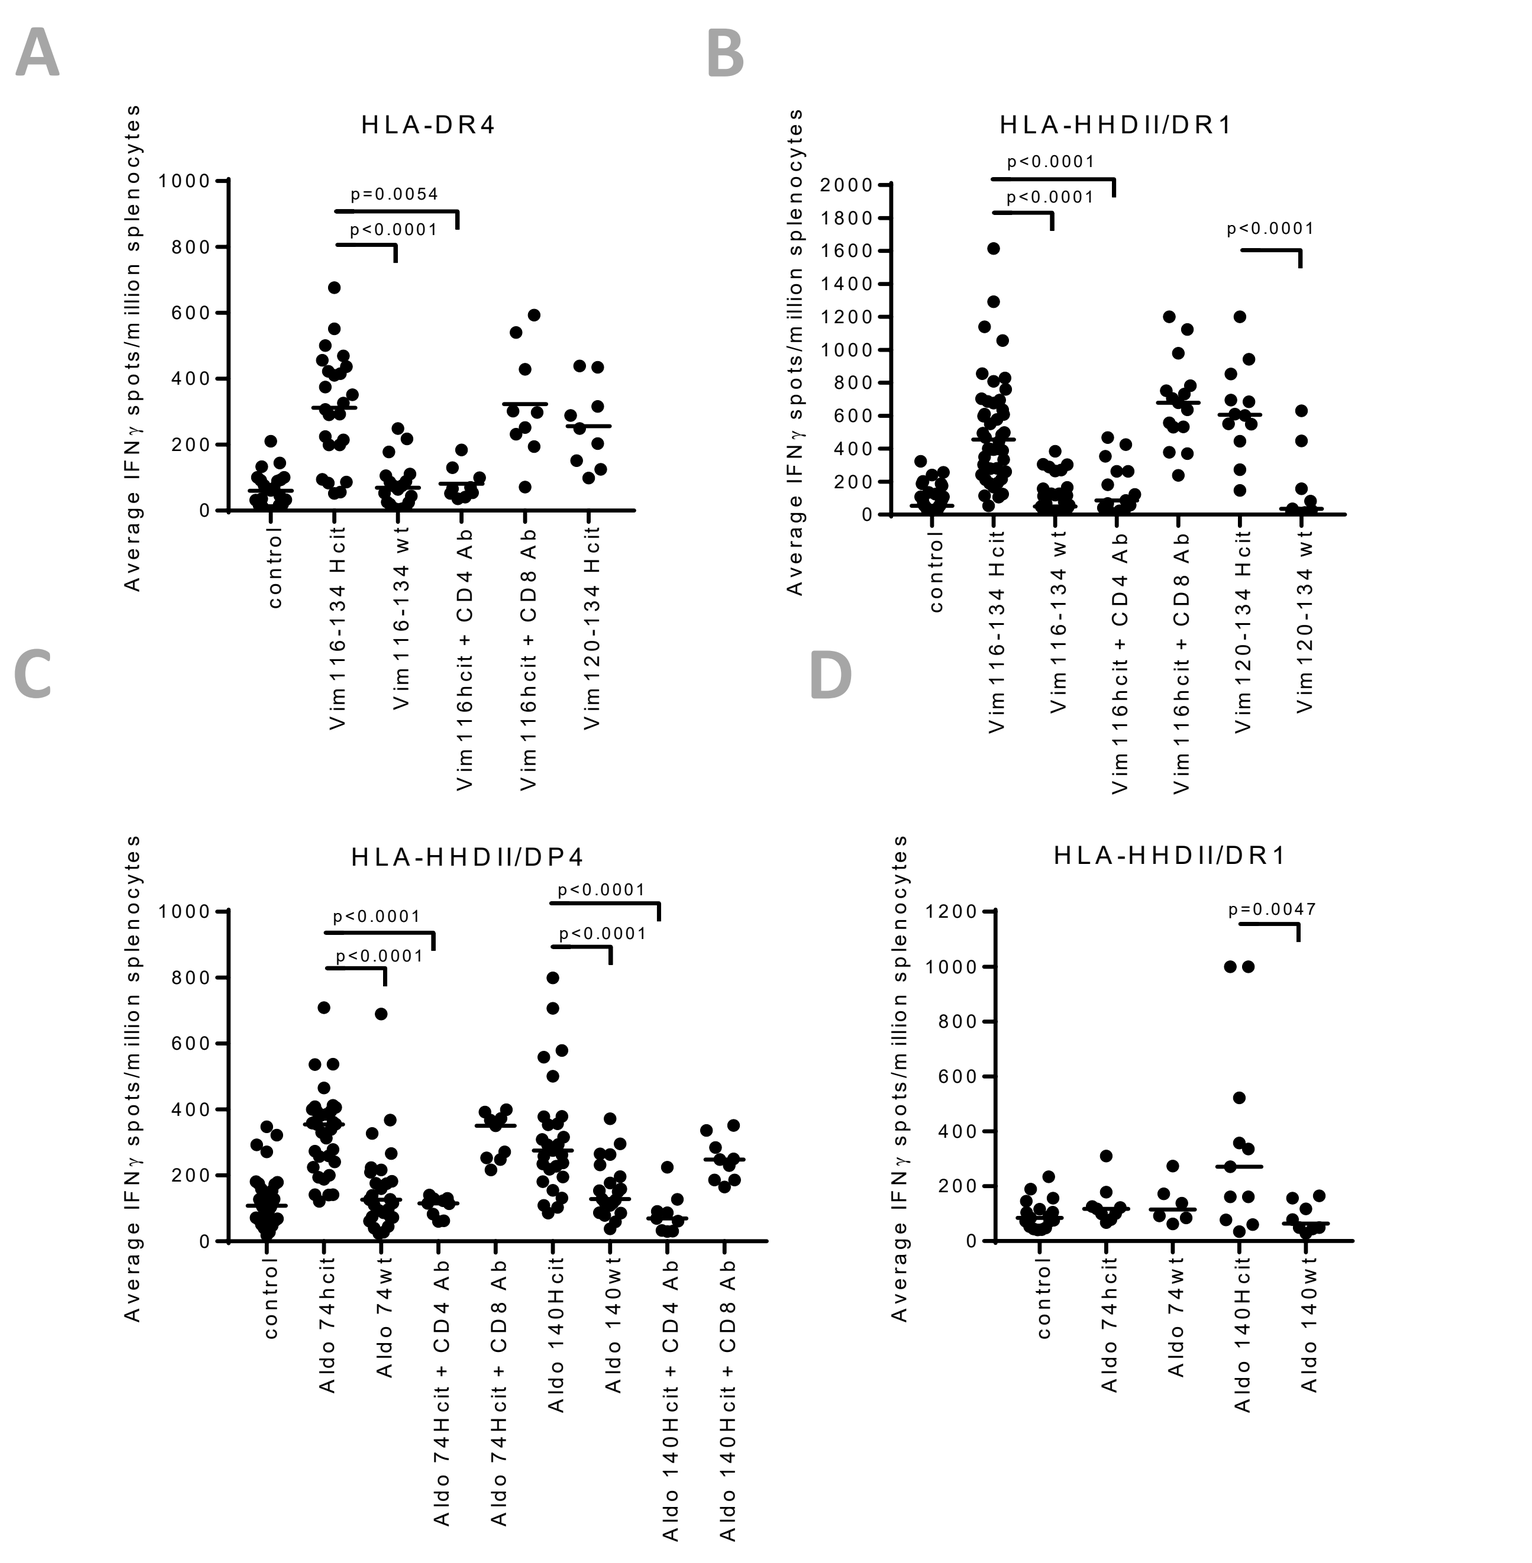
A
HLA-DR4
B
HLA-HHDII/DR1
C
HLA-HHDII/DP4
D
HLA-HHDII/DR1
Average IFNγ spots/million splenocytes

***In vivo* responses**. Immunisation with Hcit Vimentin peptides induced responses in HLA-DR4 (A) and HLA-HHDII/DR1 mice (B). Immunisation with Hcit Aldolase peptides induced responses in HLA-HHDII/DP4 (C) and HLA-HHDII/DR1 mice (D). Responses were Hcit specific and CD4 mediated for all three peptides.

Homocitrullinated peptides stimulate anti-tumour responses *in vivo*

- Mice were implanted with B16F1 tumour cells expressing human HLA on day 1 then immunised with Hcit or native (wt) peptides on day 4, 11 and 18. Tumour growth and survival was monitored.

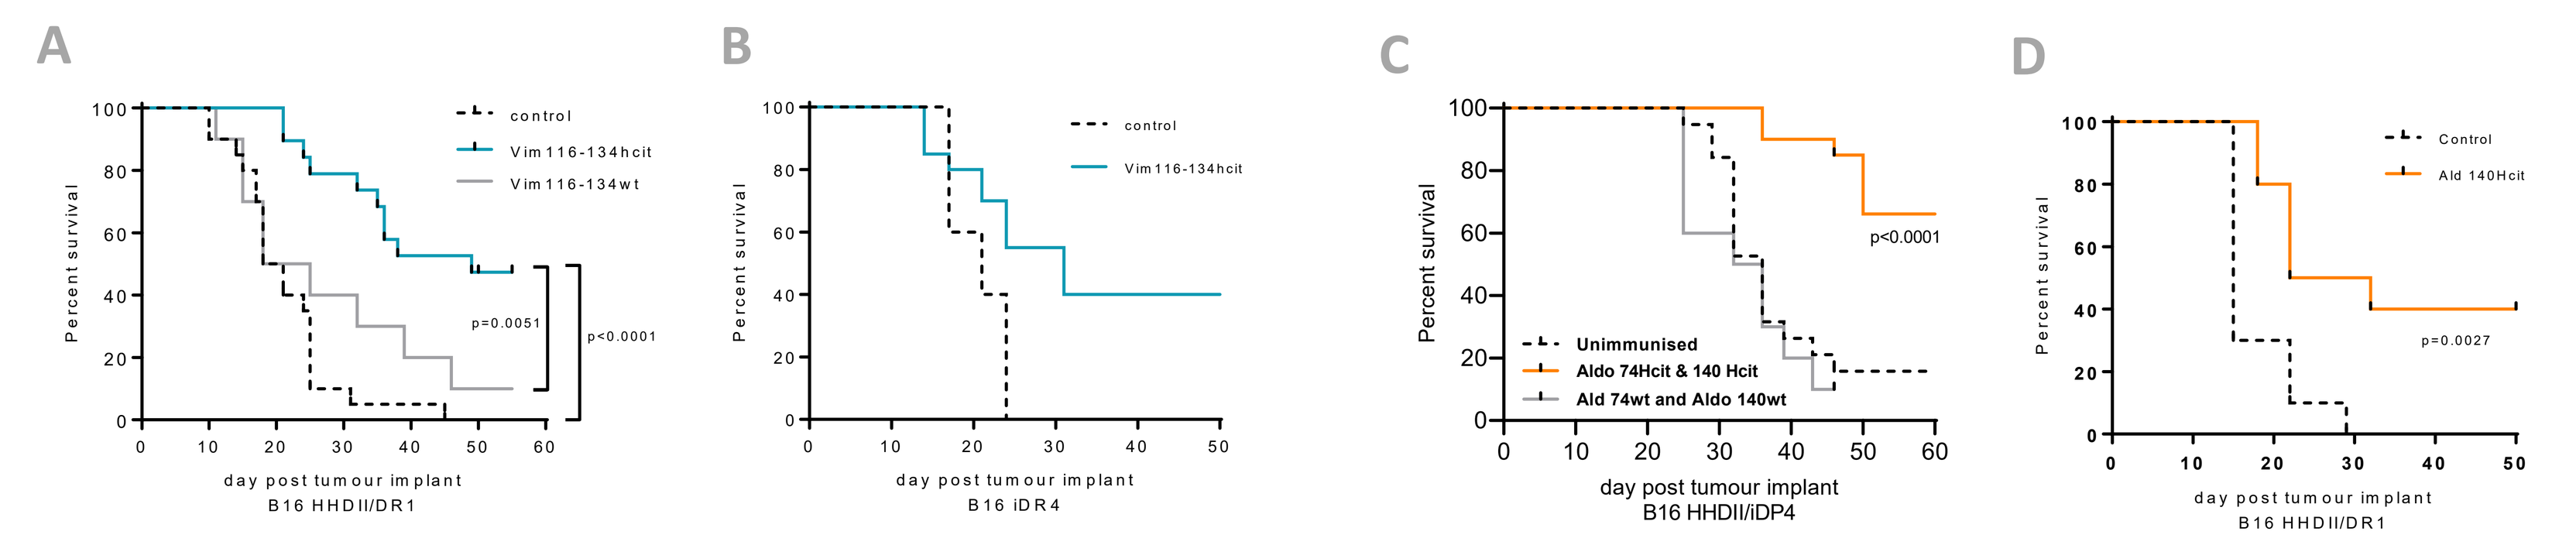
A
B
C
D
Percent survival
day post tumour implant
B16 HHDII/DR1
B16 iDR4
B16 HHDII/iDP4
p=0.0051
p<0.0001
p=0.0027

Vimentin Hcit survival. Increased survival was seen for HHDII/DR1 (A) and DR4 (B) mice immunised with the Vimentin Hcit peptide.

Aldolase Hcit survival. Increased survival when compared to control was seen for HHDII/DP4 (C) and HHDII/DR1 (D) mice immunised with Aldolase Hcit peptides.

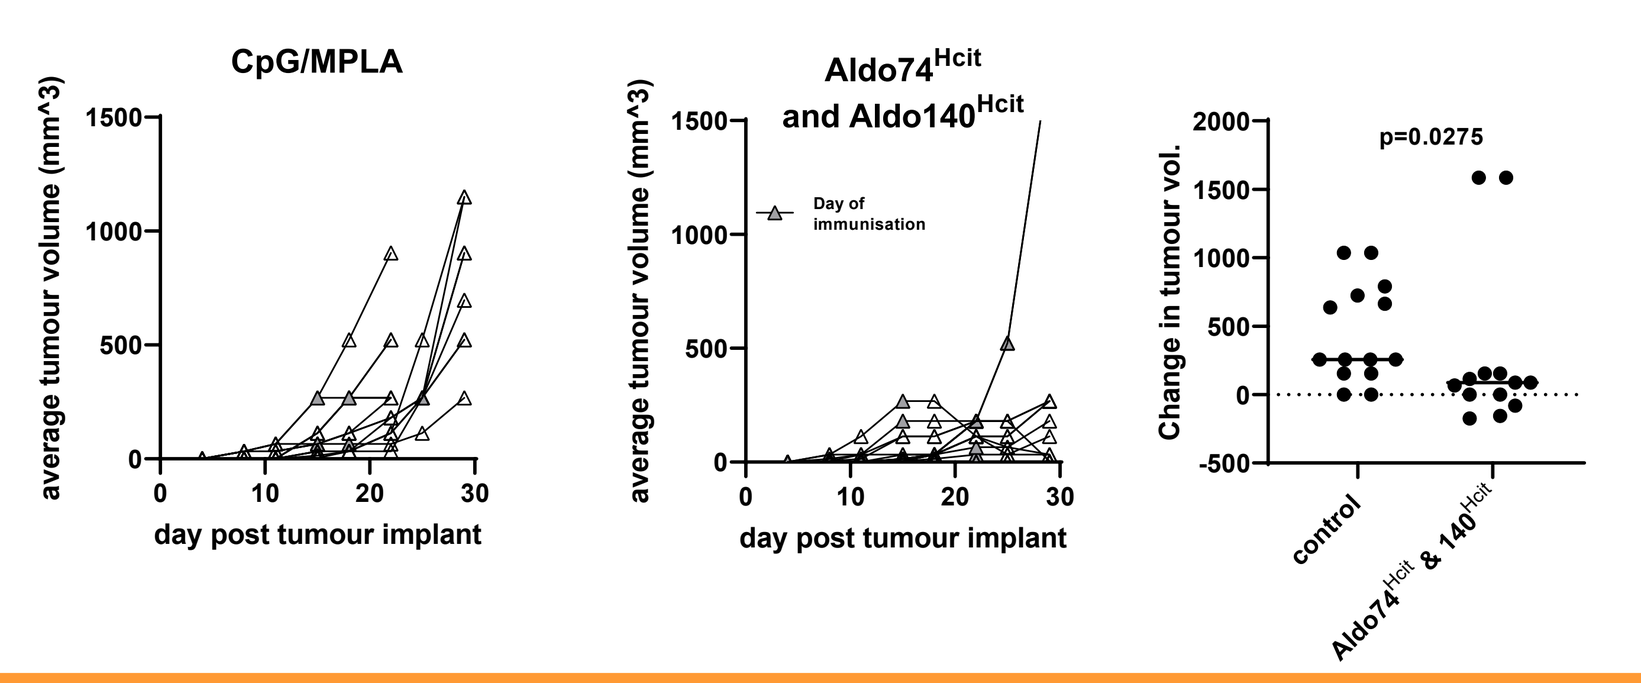
CpG/MPLA
Aldo74Hcit and Aldo140Hcit
average tumour volume (mm^3)
day post tumour implant
Change in tumour vol.
p=0.0275

Aldolase Hcit tumour regression. Immunisation with Aldolase Hcit peptides was effective against established tumours vaccinated once tumour reached a moderate size. Change in tumour volume was assessed between day of vaccination and day that mice were culled for analysis of tumour infiltrating lymphocytes.

Anti-tumour responses require MDSCs

- B16F1 tumour cells were grown *in vivo* and then dissected, disaggregated and stained for MPO expression and MDSC markers.

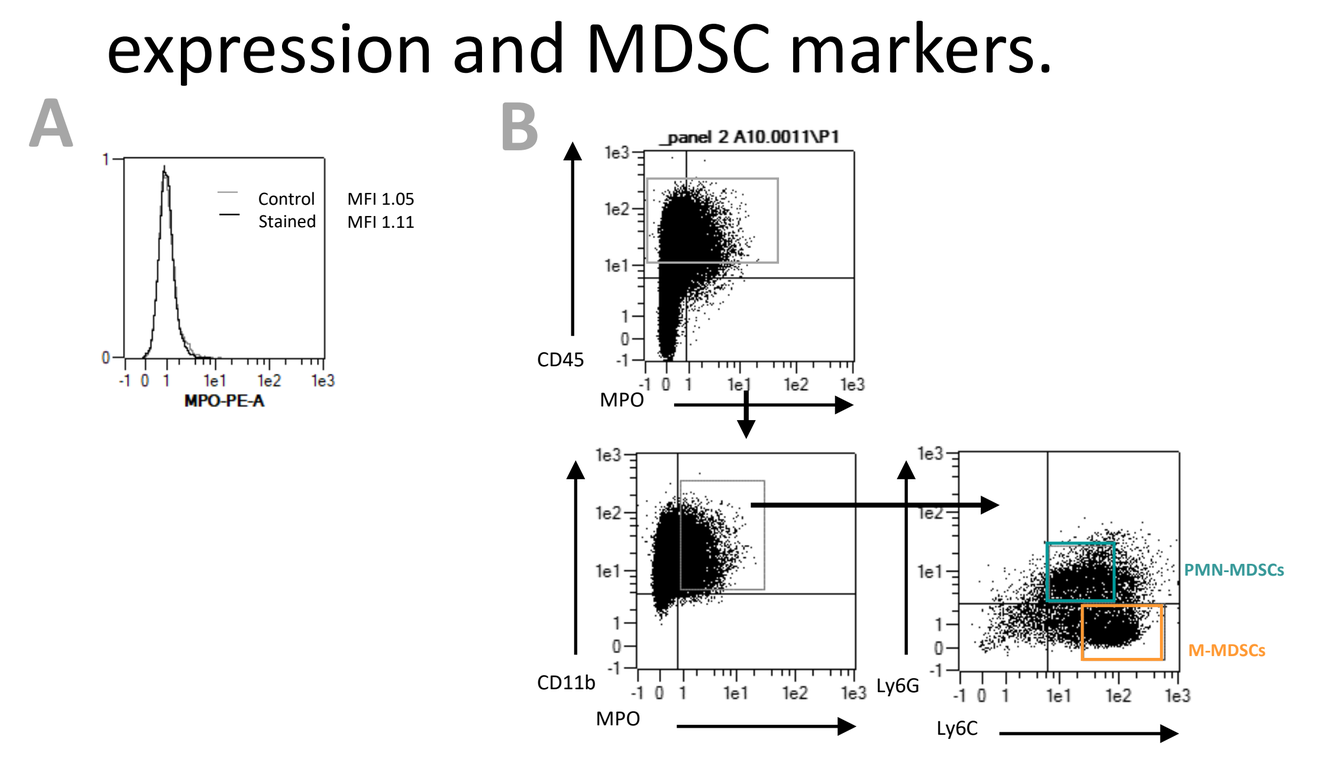
A
B
PMN-MDSCs
M-MDSCs

MPO staining. B16F1 (A) or Tumour infiltrating lymphocytes (B) were stained for MPO enzyme and immune cell markers. MPO+ cells were CD45+ CD11b+ and either Ly6G+Ly6C^{low}, markers associated with a population of polymorphonuclear MDSCs (PMN-MDSCs)/neutrophils or Ly6G-Ly6C^{hi}, markers associated with monocytic MDSCS (M-MDSCs). In the tumour both populations included MPO+ cells.

- Anti-tumour studies were carried out using Aldolase Hcit peptides in the presence of antibodies which depleted either the Ly6G+ or Ly6C+ populations.

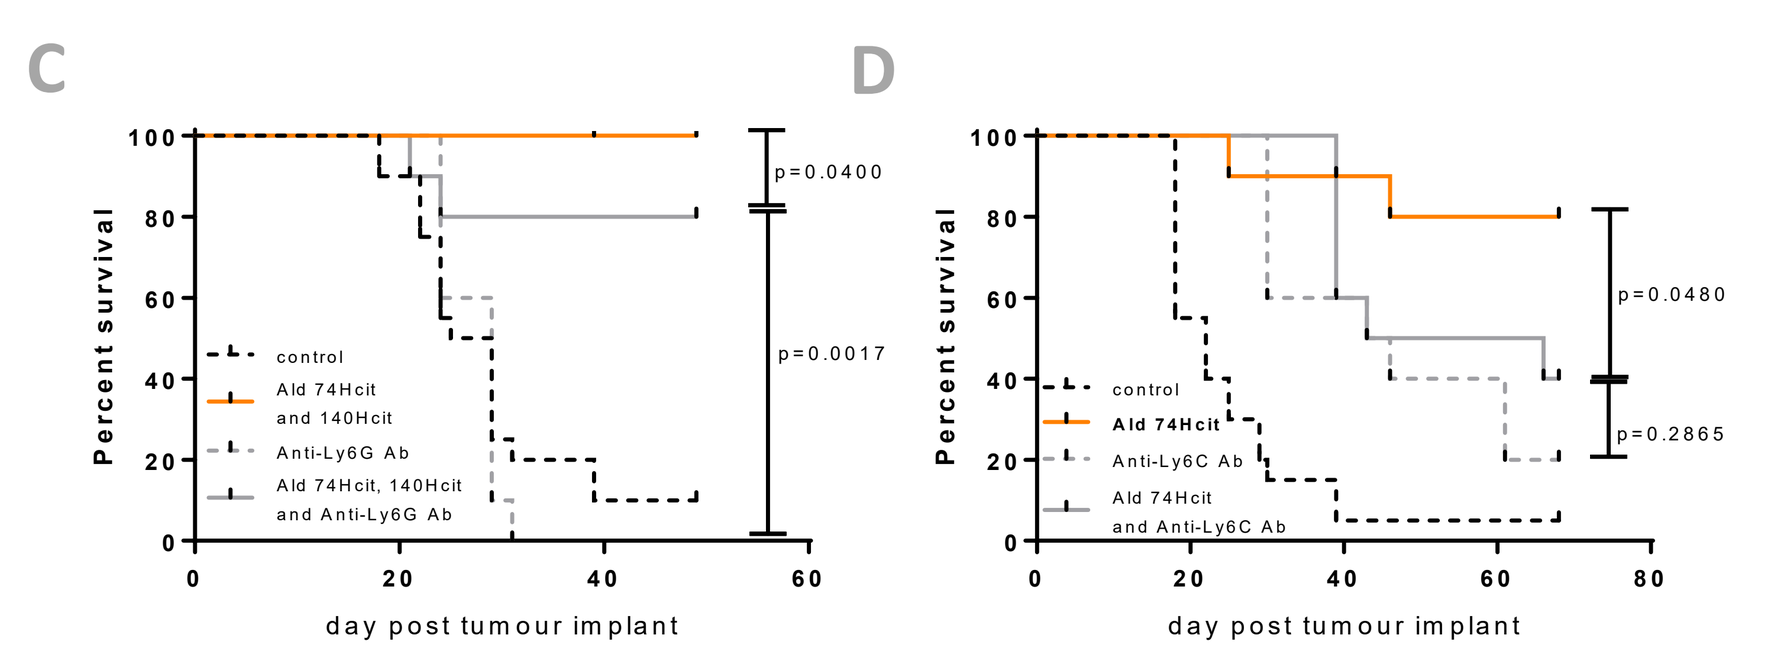
C
D
Percent survival
day post tumour implant
p=0.0400
p=0.0017
p=0.0480
p=0.2865

Survival in Ly6G or Ly6C depleted mice. HHDII/DP4 mice immunised with Aldolase Hcit peptides in the presence of antiLy6G antibody showed little decrease in survival (C). In contrast mice immunised with Aldolase Hcit peptides in the presence of anti-Ly6C antibodies showed reduced survival compared to Hcit peptides only (D).

Anti-tumour responses require CD4 T cells and MHCII presentation

- Anti-tumour studies were also carried out in the presence of antibodies which depleted either CD4 or CD8 cell populations.

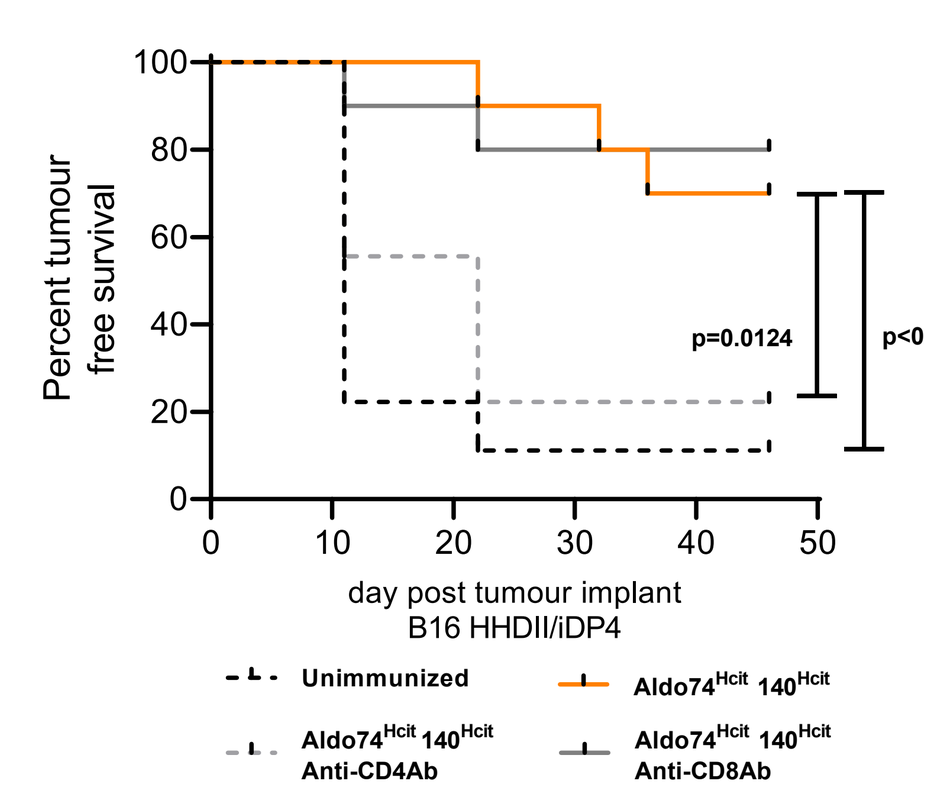
A
Percent tumour free survival
day post tumour implant
B16 HHDII/iDP4
p=0.0124
p<0.0001

Survival in CD4/CD8 depleted mice. Immunisation with Aldolase 74Hcit and Aldolase 140Hcit peptides had an anti-tumour effect in CD8 depleted mice but not in CD4 depleted mice (A)

- Mice were implanted with B16F1 cells lacking MHC II expression.
- These mice have MHCII presentation on APCs but not on implanted tumour cells.

Survival in MHC II deficient model. Immunisation with Aldolase 74Hcit and 140Hcit peptides had no effect on survival in HHDII/DP4 mice challenged with B16F1 cells without MHC II expression (B).

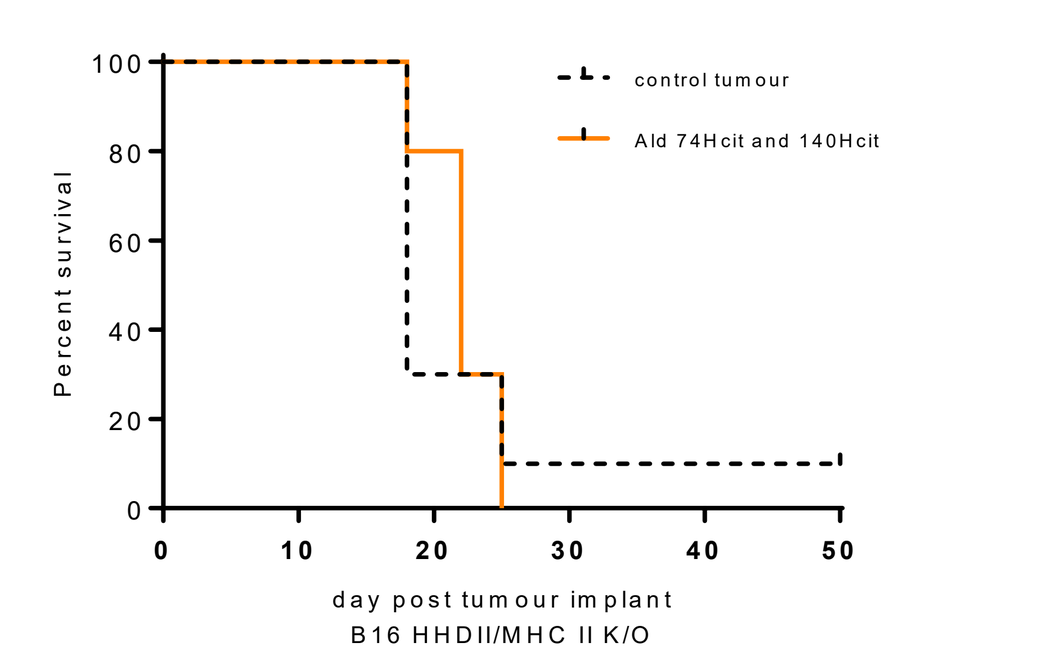
B
Percent survival
day post tumour implant
B16 HHDII/MHC II K/O

Humans have a repertoire for homocitrullinated peptides

- PBMCs were isolated from 15 healthy donors and 11 cancer (LG – lung, OV – ovarian, BR – breast) patients.
- PBMCs were CD25 depleted and CFSE labelled then stimulated with each peptide for 10 days.

Proliferative responses. Proliferation was measured in the stimulated samples from each donor. Most donors showed CD4 proliferation in response to at least one of the Homocitrullinated peptides in both the healthy donors (A) and patients (B)

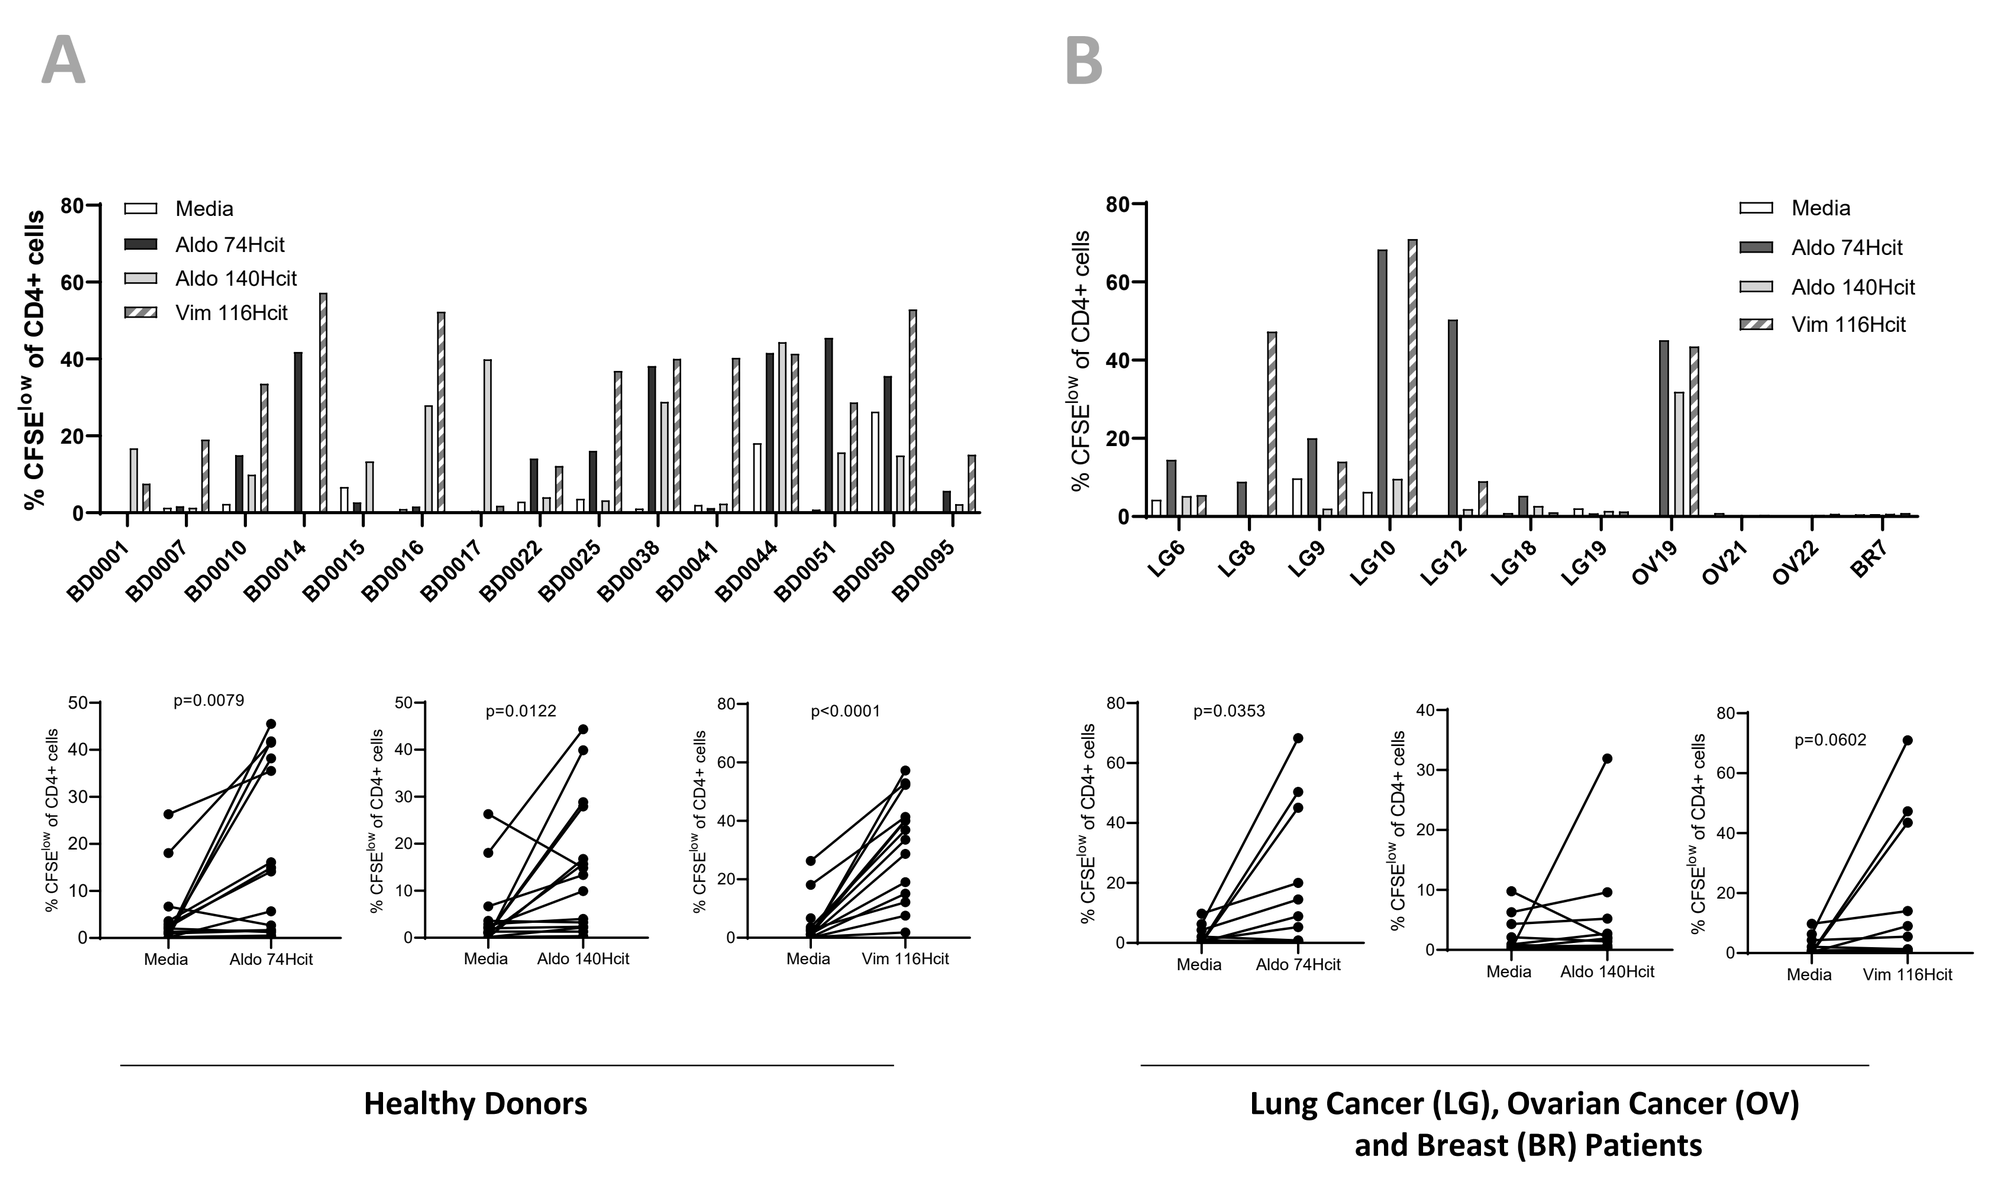
A
B
Healthy Donors
Lung Cancer (LG), Ovarian Cancer (OV) and Breast (BR) Patients

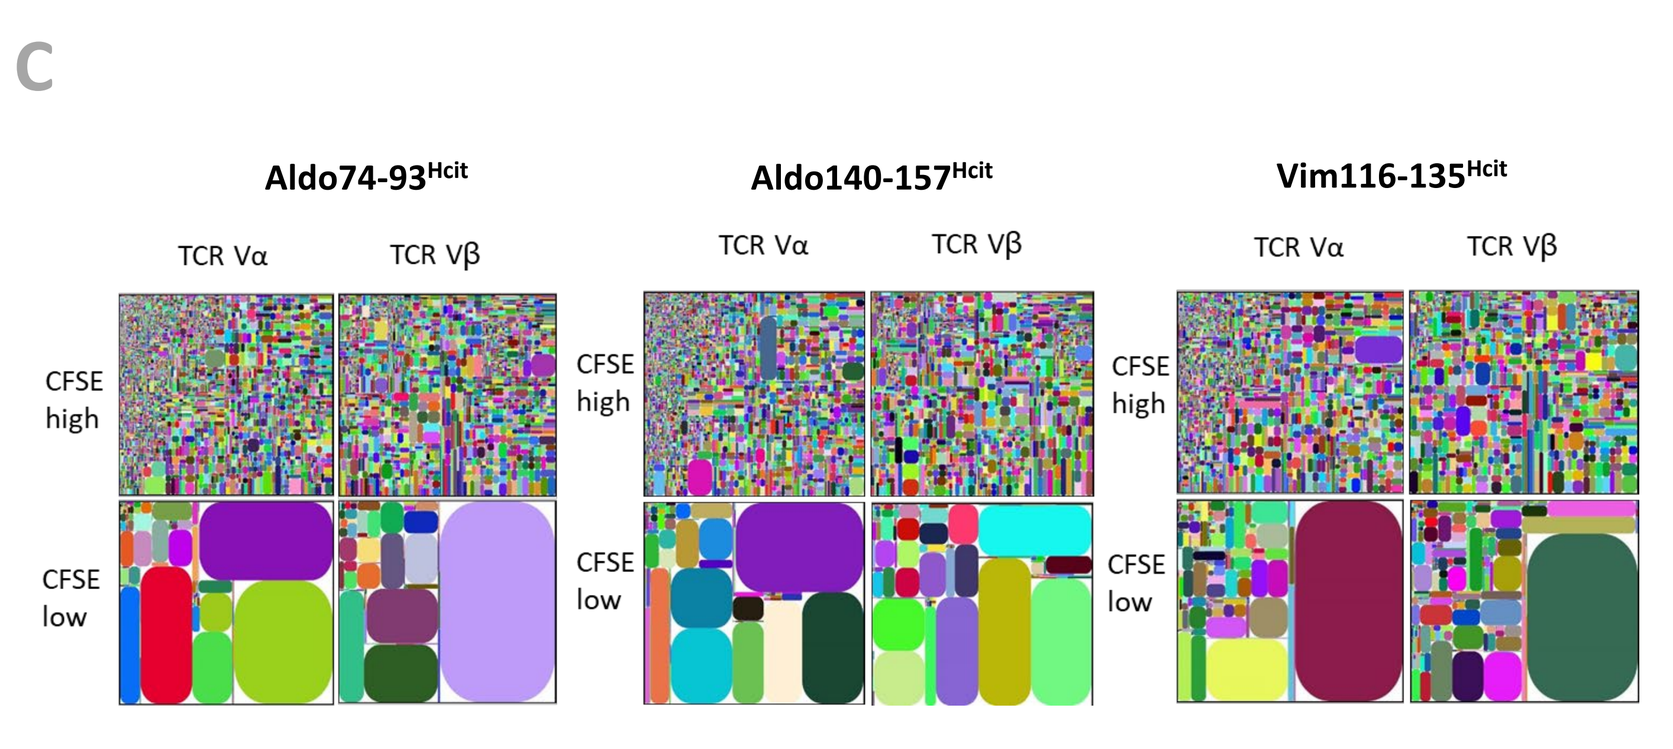
C
Aldo74-93Hcit
Aldo140-157Hcit
Vim116-135Hcit
TCR Vα
TCR Vβ
CFSE high
CFSE low

TCR analysis of proliferating population. The CD4+CFSE^{low} and CFSE^{high} cells from responding donors were sorted by flow cytometry and TCR analysis was performed. In the proliferating (CFSE^{low}) population the responses are oligoclonal compared to the non-proliferating (CFSE^{high}) population (C)

CONCLUSIONS

Immunisation with Homocitrullinated peptides induces strong CD4 responses

- **Homocitrullinated Vimentin and Aldolase peptides can induce CD4 T-cell responses restricted via a number of HLA types**
- **Responses after Hcit peptide vaccination do not cross react with wild type, non-homocitrullinated peptides and are CD4 mediated**

Hcit peptide responses lead to increased survival *in vivo* This seems to involve MPO production by MDSCs leading to increased carbamylation in the tumour microenvironment and MHC II presentation of homocitrullinated epitopes

- **Hcit vaccination is associated with increased survival *in vivo***
- **Responses are attenuated in mice which have been depleted of Ly6C^{high} cells which includes MPO producing MDSCs**
- **However, responses also require tumour cells to have the ability to express MHC II and are mediated by CD4 cells**

Human donors also showed a repertoire of CD4 T-cells that respond to these peptides

1. Brentville VA, Metheringham RL, Gunn B, Symonds P, Daniels I, Gijon M, Cook K, Xue W, Durrant LG (2016). Citrullinated vimentin presented on MHC-II in tumor cells is a target for CD4+ T cell-mediated antitumor immunity. Cancer Research 2016 Feb 1;76(3):548-60
2. Cook K, Daniels I, Symonds P, Pitt T, Gijon M, Xue W, Metheringham R, Durrant L, Brentville V. Citrullinated α-enolase is an effective target for anti-cancer immunity. Oncoimmunology. 2017 Nov 6;7(2):e1390642
3. Holzer, M., K. Zangger, D. El-Gamal, V. Binder, S. Curcic, V. Konya, R. Schuligoi, A. Heinemann, and G. Marsche. 2012. 'Myeloperoxidase-derived chlorinating species induce protein carbamylation through decomposition of thiocyanate and urea: novel pathways generating dysfunctional high-density lipoprotein', Antioxid Redox Signal, 17: 1043-52.
4. Jaisson, S., C. Pietrement, and P. Gillery. 2011. 'Carbamylation-derived products: bioactive compounds and potential biomarkers in chronic renal failure and atherosclerosis', Clin Chem, 57: 1499-505.

PRINTED BY SCIENCEPOSTERS .co.uk