Targeting gastrointestinal tumors with constant region - engineered anti - glycan antibodies

Mireille Vankemmelbeke[1], T Kirk[1], C Papagregoriou[2], S Aziz[2], JX Chua[1], RS McIntosh[2], LG Durrant[1,2]
[1]Scancell Holdings plc, Oxford UK, [2]Nottingham University, Nottingham, UK

INTRODUCTION

- avid antibody (mAb) binding to cell surface glycotargets leads to direct tumor cell killing in the absence of immune effector cells or complement[1] (A)
- this direct cell killing is isotype-restricted to mouse IgG3 (mIgG3), which exhibit cooperative target binding[2]
- we hypothesize that the mAb-induced direct cell killing is a form of inflammatory/immunogenic cell death (ICD) with the potential to initiate an adaptive anti-tumor immune response, leading to long-term tumor control (B) [3]
- we engineered human IgG1 mAbs with direct cell killing ability through constant region modification

A

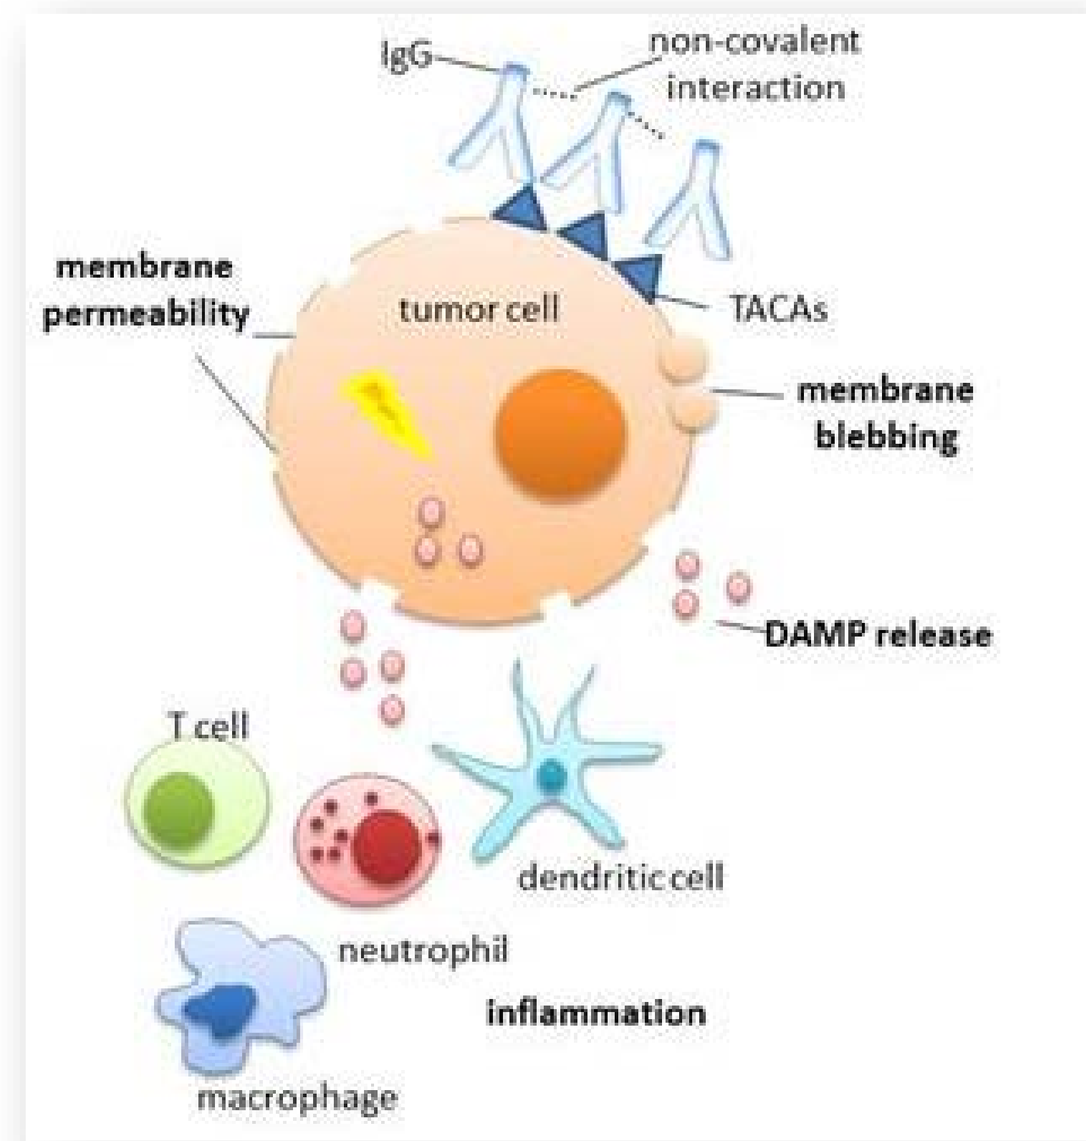
IgG
non-covalent interaction
membrane permeability
tumor cell
TACAs
membrane blebbing
DAMP release
T cell
dendritic cell
neutrophil
inflammation
macrophage

B

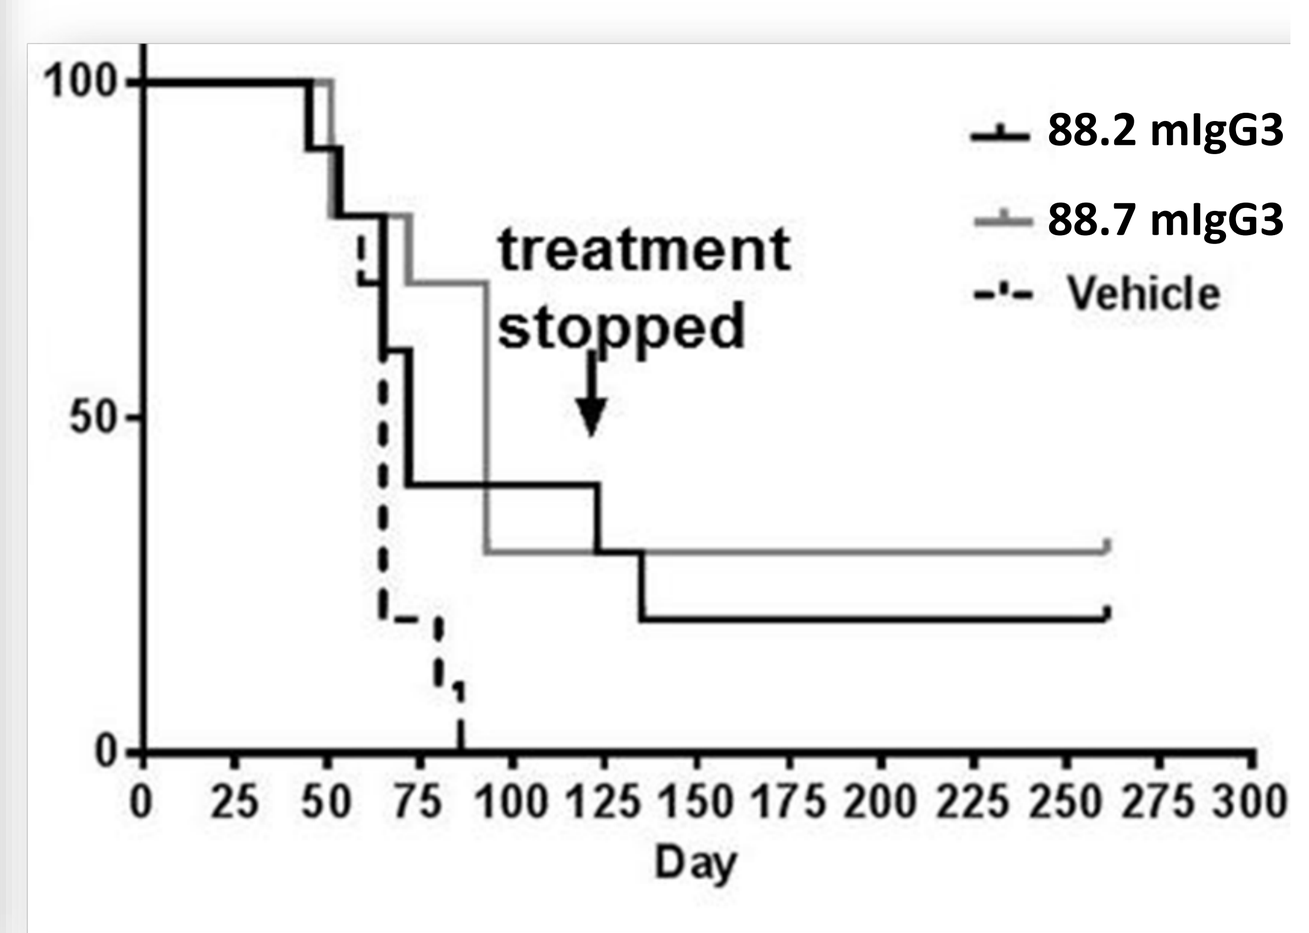
88.2 mIgG3
88.7 mIgG3
Vehicle
treatment stopped
Day

CONCLUSIONS

- **intermolecular cooperativity underlies mIgG3 isotype restriction of anti-glycan mAbs in mice**
- **tumor glycan-binding mIgG3 elicit direct cell killing, aided by avid target binding, a phenomenon not maintained in hIgG1 - this direct cell killing exhibits ICD features**
- **we have generated a panel of direct cell killing human 'iG1' mAbs via the introduction of selected mIgG3 residues into hIgG1 glycan-binding mAbs**
- **i129G1 with improved direct tumor cell killing is a prime candidate for therapeutic development**

avid glycan binding by 88mIgG3, but not 88hIgG1

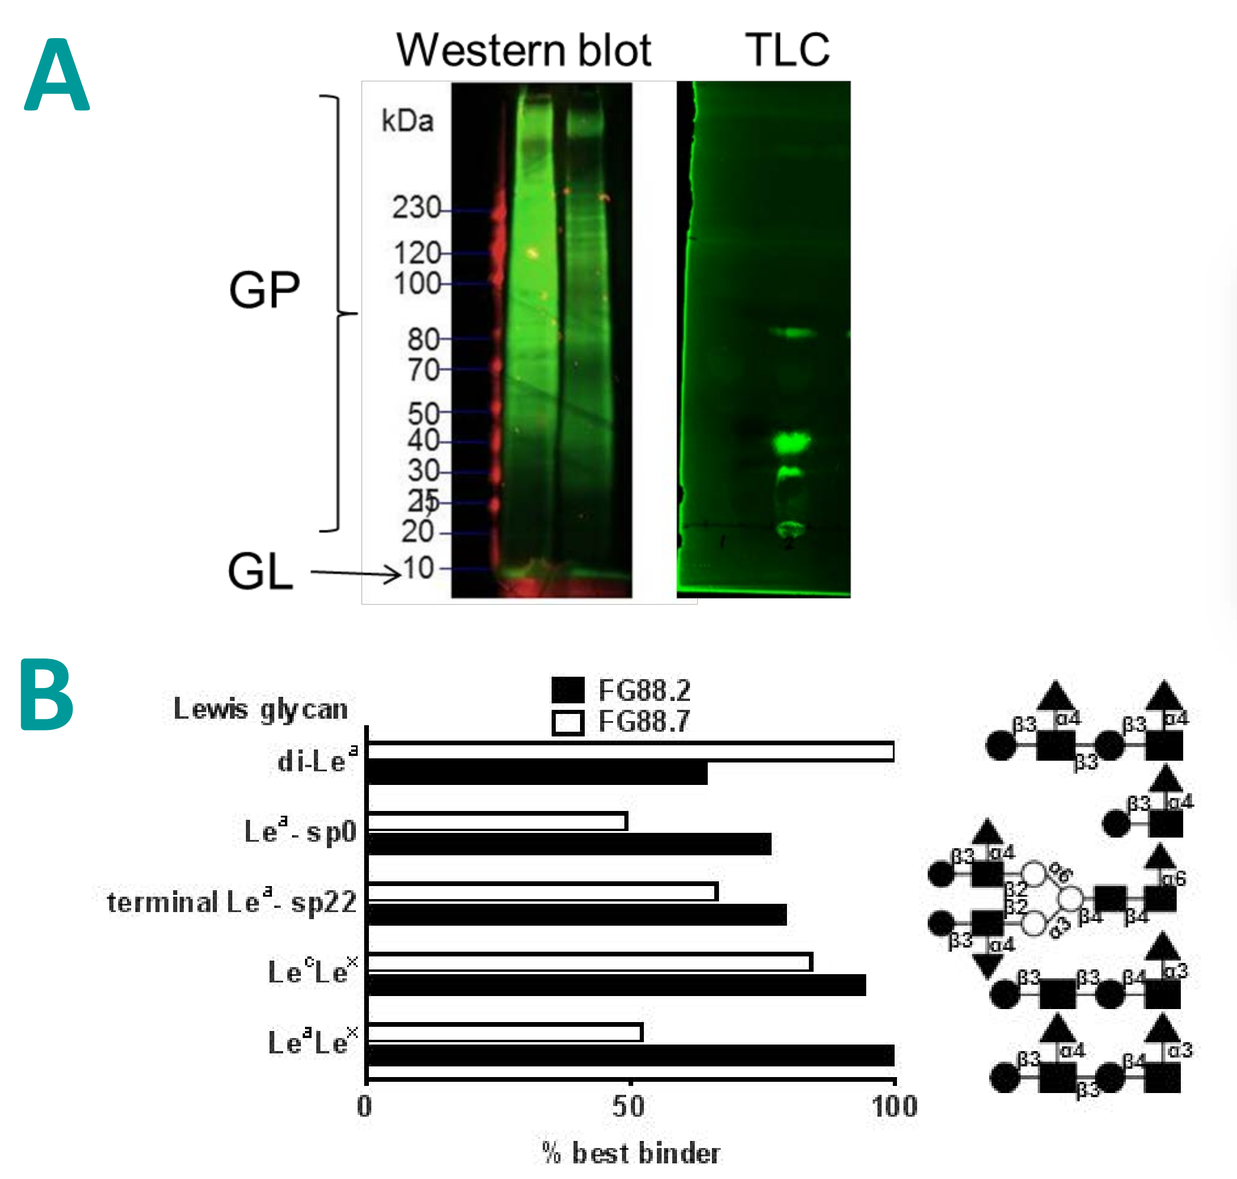
A
Western blot
TLC
kDa
GP
GL
B
Lewis glycan
FG88.2
FG88.7
% best binder

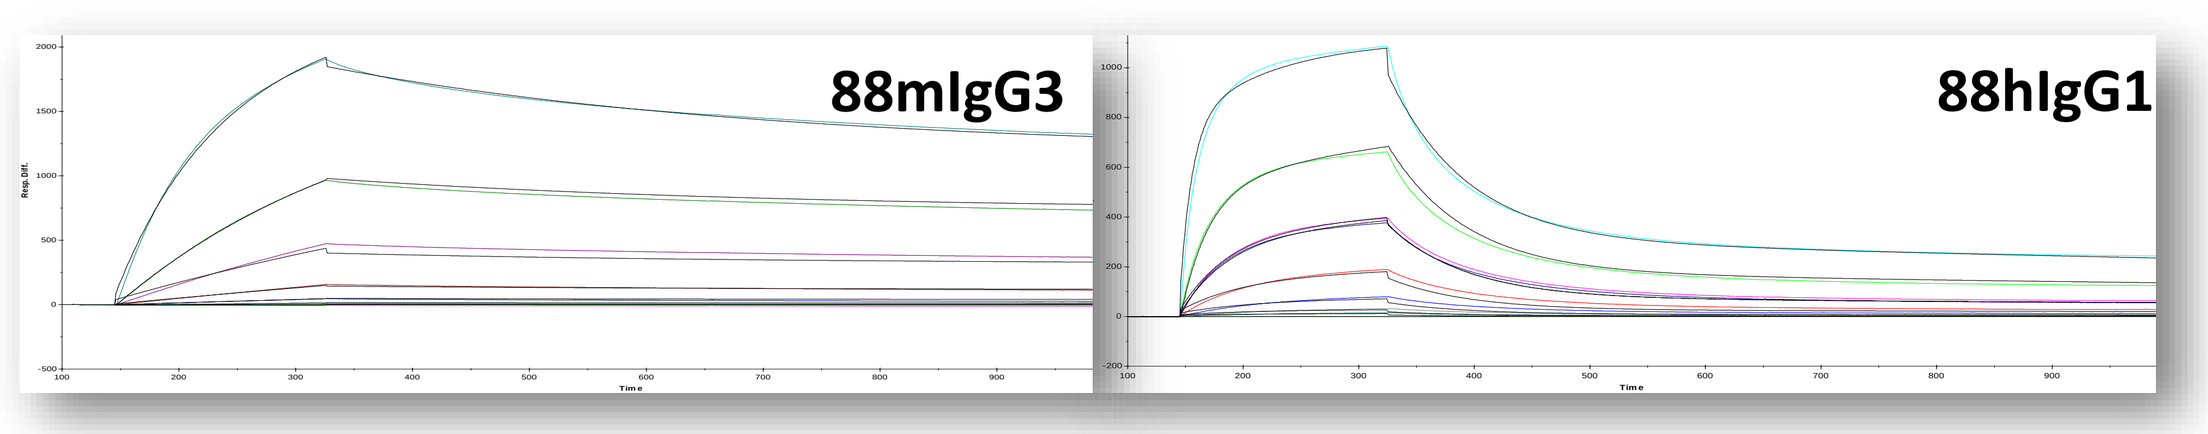
C
88mIgG3
88hIgG1

D

mAb	Association Rate k_{on} (1/Ms)	Dissociation Rate k_{off} (1/s)	Kd (M)
88mIgG3	7.0 x 10^{4}	3.6 x 10^{-6}	5.2 x 10^{-11}
88hIgG1	6.1 x 10^{4}	4.1 x 10^{-4}	6.7 x 10^{-9}

Figure 1.

western blot analysis and thin layer chromatography of 88mIgG3 binding to CRC's plasma membrane lipid extract (A). The mAb binding is driven by Lewis^{a,c,x} recognition (B). Real-time Lewis^{a} binding by 88mIgG3 and 88hIgG1 (Biacore 3000) (C), kinetic binding constants derived from fitting titration series of mAb to a heterogenous ligand binding model (D). 88mIgG3 displays high glycan functional affinity, in contrast to 88hIgG1

high-level 88mIgG3 cancer cell binding induces cellular aggregation, pore formation and cell death

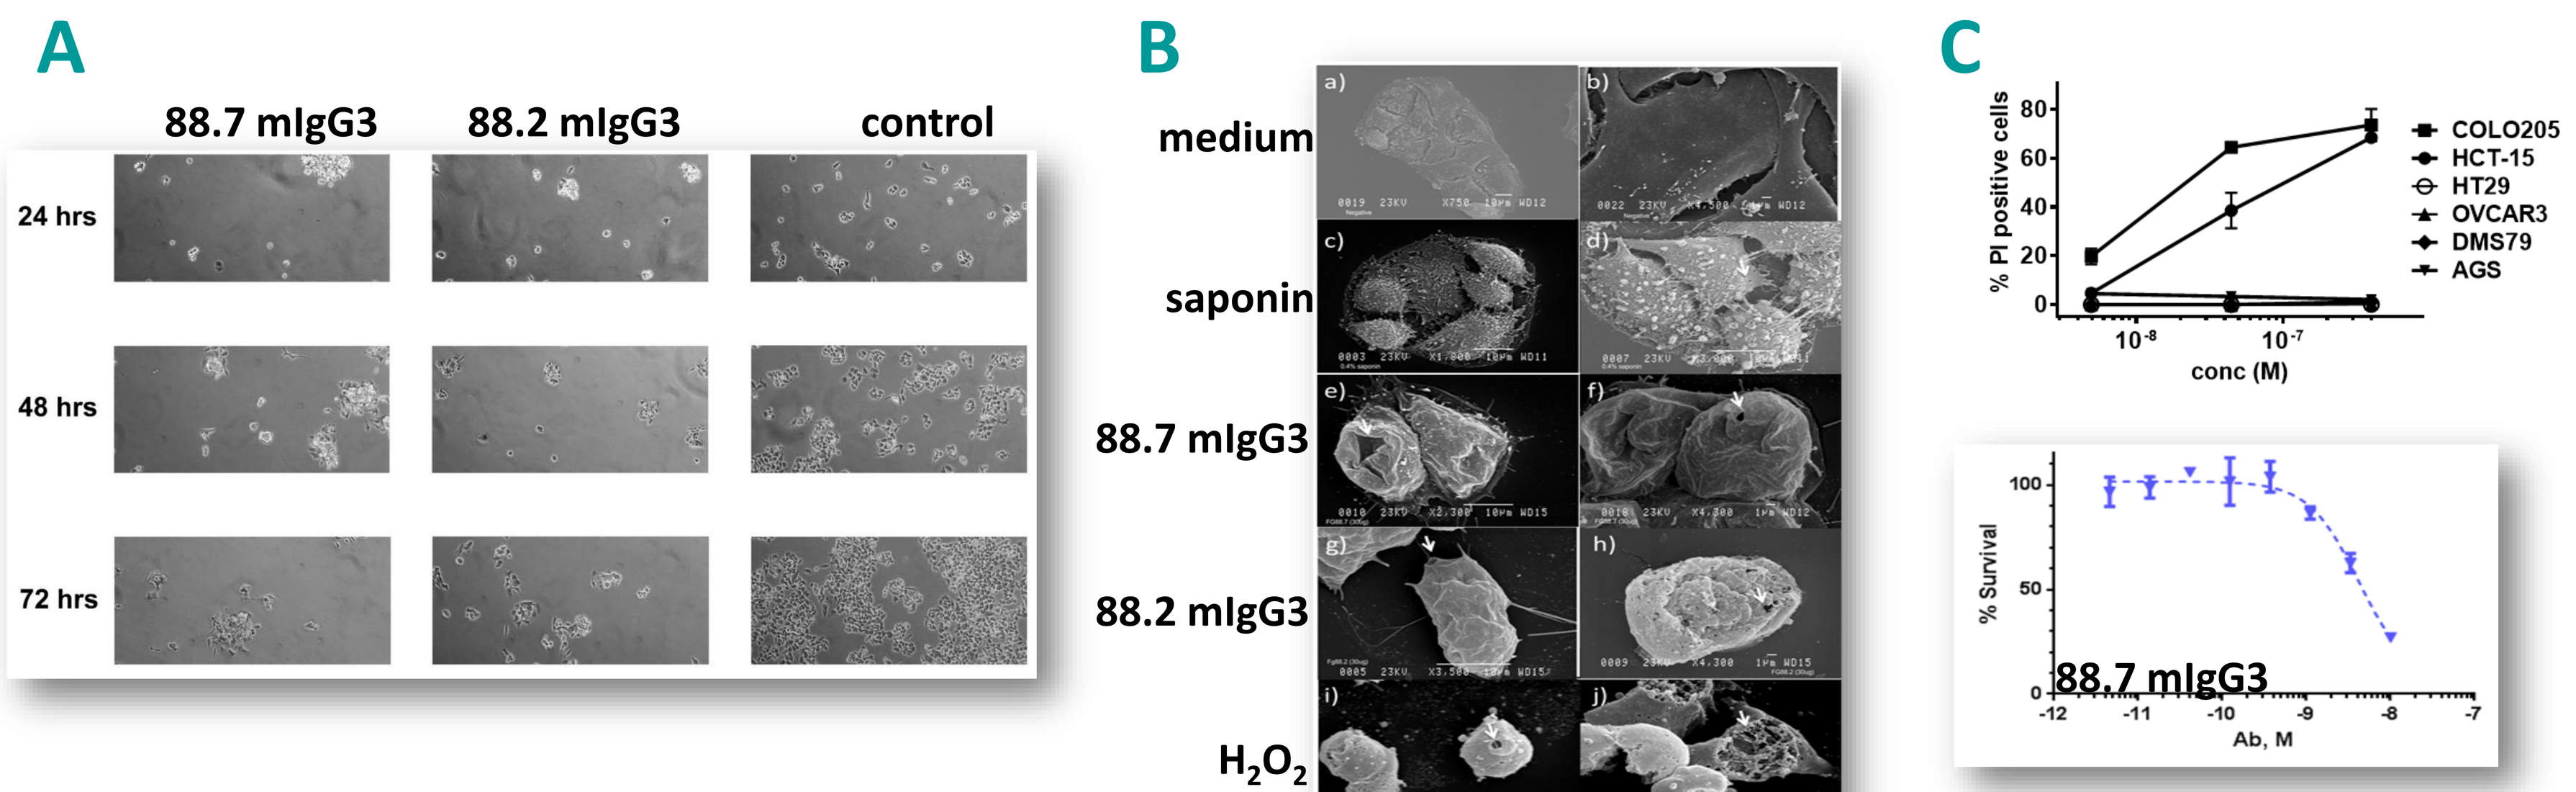
A
88.7 mIgG3
88.2 mIgG3
control
24 hrs
48 hrs
72 hrs
B
medium
saponin
88.7 mIgG3
88.2 mIgG3
H2O2
C
% PI positive cells
conc (M)
COLO205
HCT-15
HT29
OVCAR3
DMS79
AGS
% Survival
88.7 mIgG3
Ab, M

Figure 2.

light microscopy showing cellular aggregation of 88mIgG3-treated HCT15 cells (A); scanning electron microscopy (SEM) of 88mIgG3-treated HCT15 cells (arrows point to heterogeneous pores (B); 88mIgG3-mediated cytotoxicity: top, propidium iodide uptake on high-binding colorectal cancer cell lines (CRC), but not on low-binding CRC lines; bottom, 88mIgG3-mediated inhibition of proliferation of HCT15 cells (IC_{50}: 10^{-9}M) (C)

i129G1, an anti-sialylated Lewisa glycan mAb with improved in vitro and in vivo direct cell killing

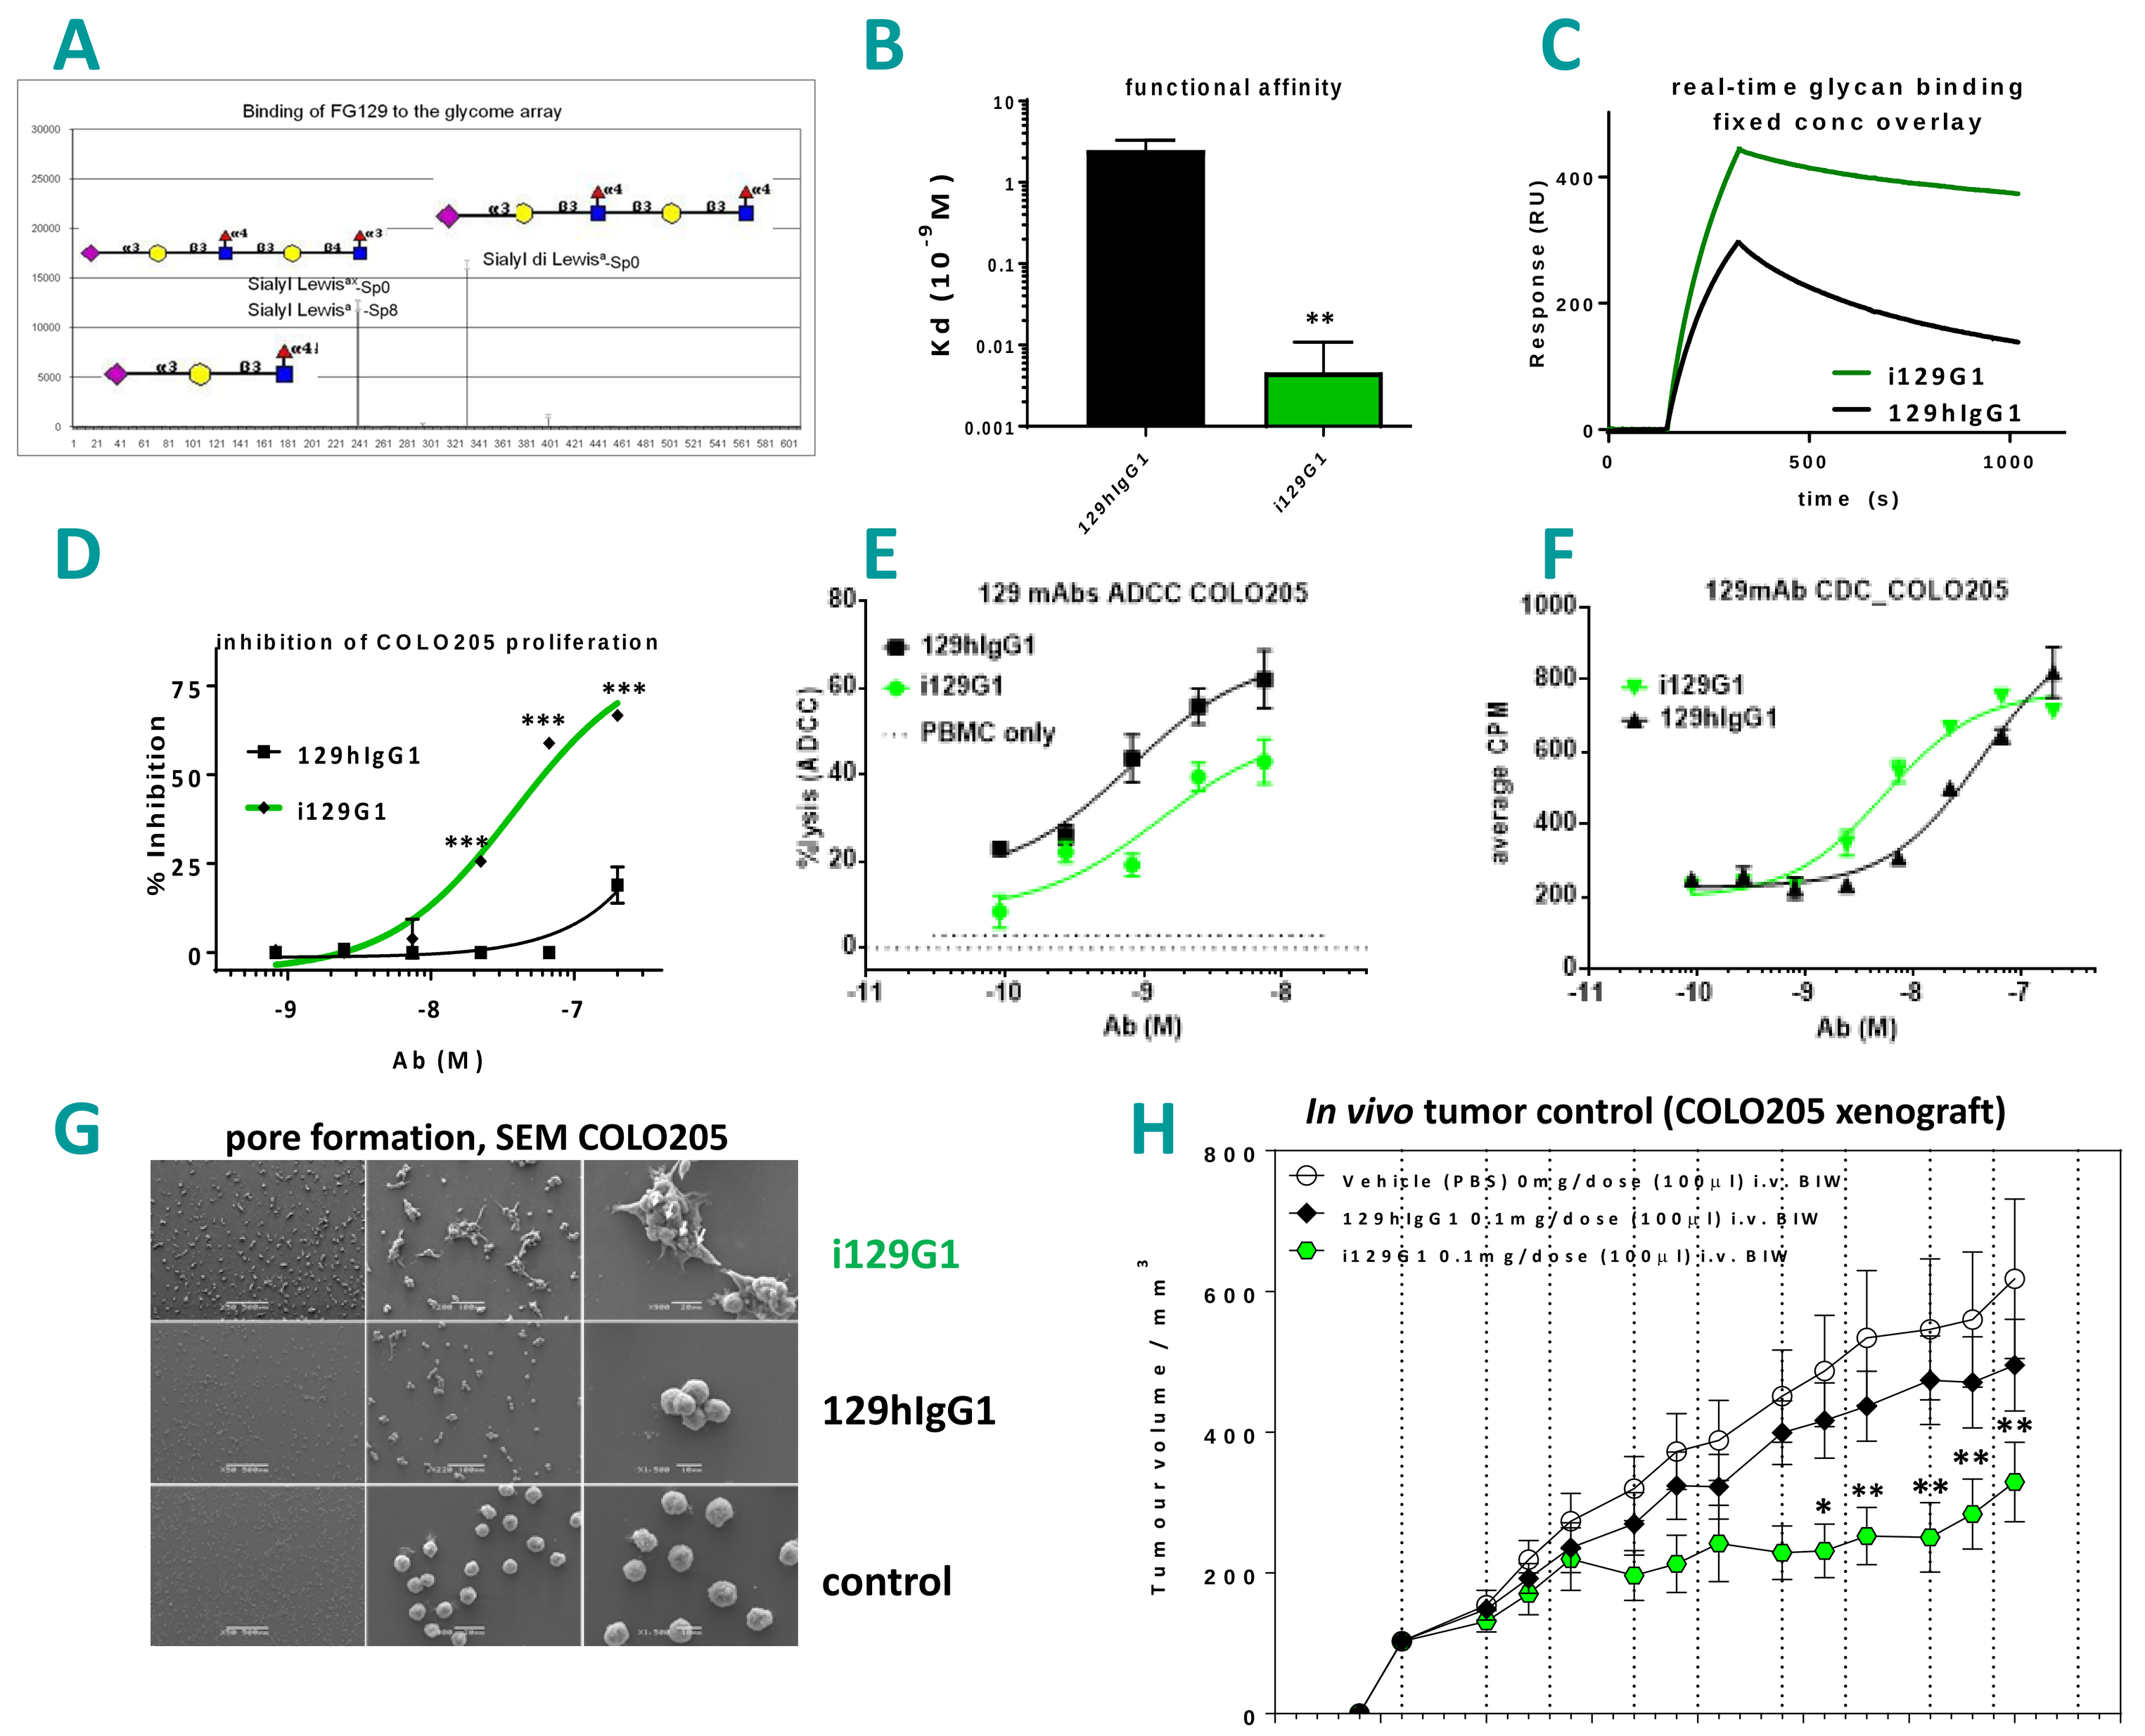
A
Binding of FG129 to the glycome array
Sialyl Lewisax-Sp0
Sialyl Lewisa -Sp8
Sialyl di Lewisa-Sp0
B
functional affinity
Kd (10-9 M)
129hIgG1
i129G1
C
real-time glycan binding
fixed conc overlay
Response (RU)
time (s)
D
inhibition of COLO205 proliferation
% Inhibition
Ab (M)
E
129 mAbs ADCC COLO205
PBMC only
%lysis (ADCC)
F
129mAb CDC_COLO205
average CPM
G
pore formation, SEM COLO205
i129G1
129hIgG1
control
H
In vivo tumor control (COLO205 xenograft)
Vehicle (PBS) 0mg/dose (100µl) i.v. BIW
129hIgG1 0.1mg/dose (100µl) i.v. BIW
i129G1 0.1mg/dose (100µl) i.v. BIW
Tumour volume / mm3
Study day

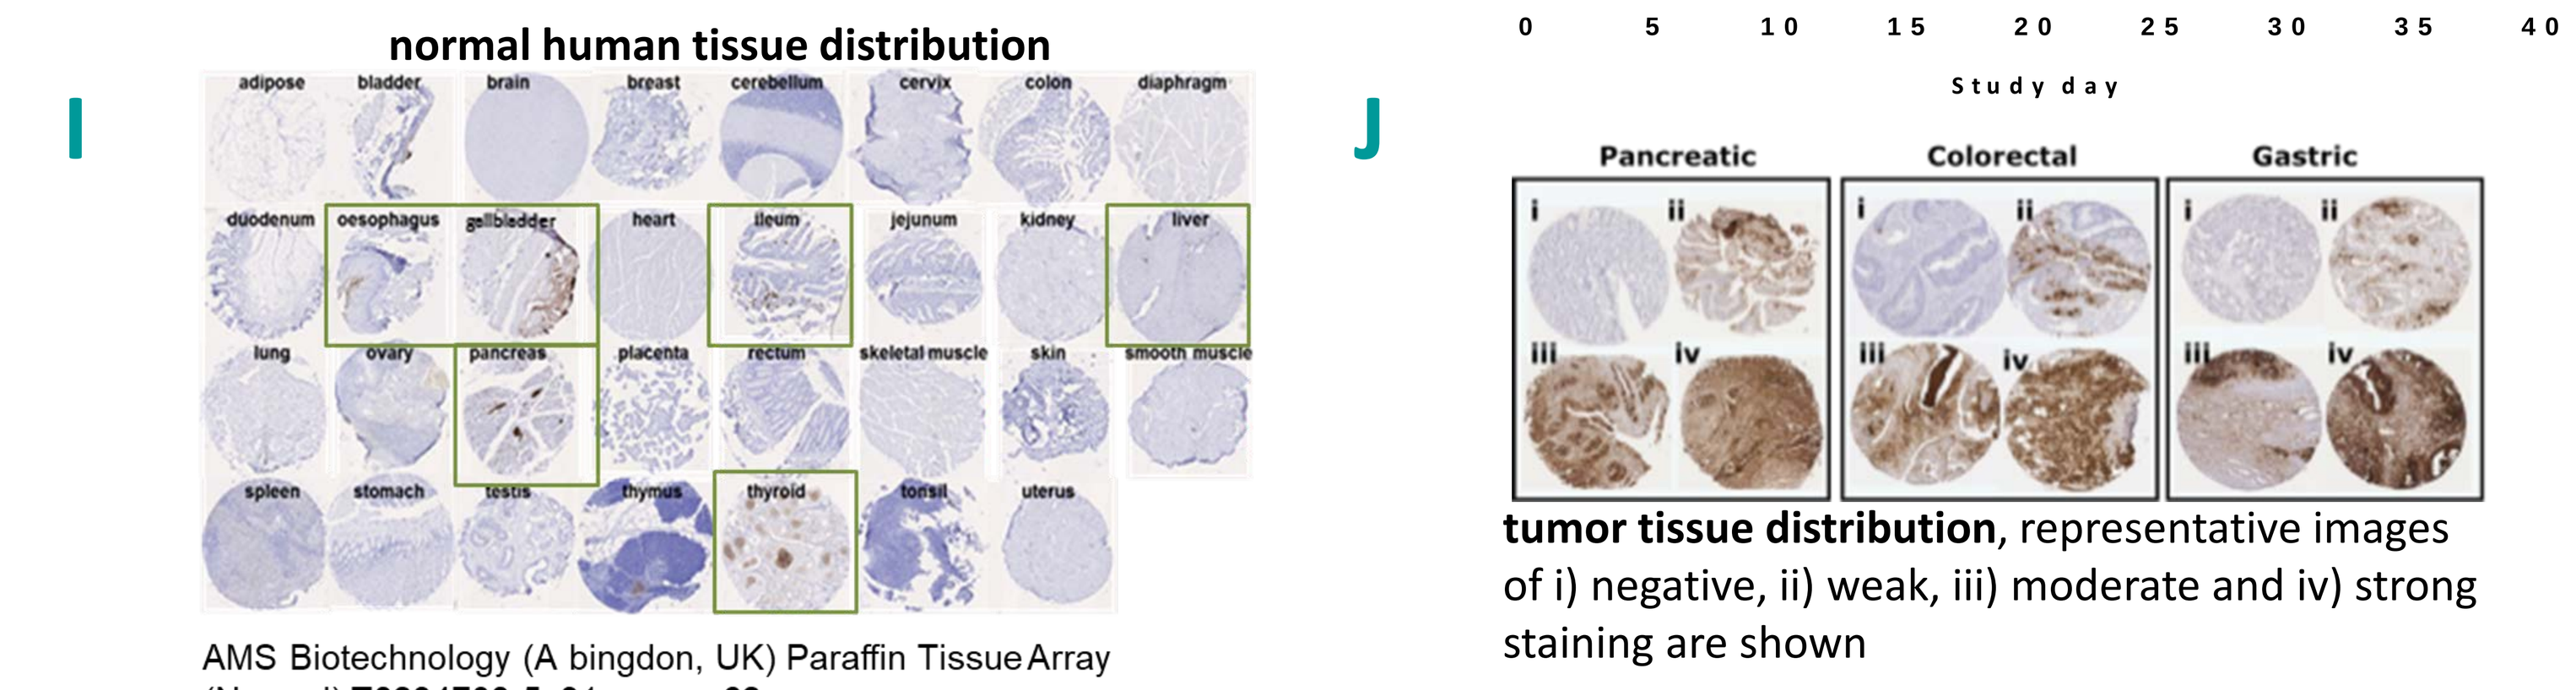
I
normal human tissue distribution
J
Pancreatic
Colorectal
Gastric

AMS Biotechnology (A bingdon, UK) Paraffin Tissue Array (Normal) T8234708-5, 31 cases, 62 cores

tumor tissue distribution, representative images of i) negative, ii) weak, iii) moderate and iv) strong staining are shown

Figure 6.

- 129hIgG1 selectively binds sialylated (di)Lewis^{a} -containing glycan (A); introduction of mIgG3 constant region residues into 129hIgG1, creates an improved i129G1 with significantly improved glycan functional affinity, resulting from a significantly slower off-rate (B, C) and direct tumor cell killing (COLO205) (D). i129G1 maintains ADCC on COLO205 (E) and exhibits improved CDC activity on HCT15 (F). i129G1 induces cellular aggregation and pore formation on COLO205 (G).
- i129G1 exhibits significant tumor volume reduction in a COLO205 xenograft model (H). (BALB/c nude mice, CrownBio). Dosing commenced six days after tumor inoculation (100mm^{3} mean tumor volume). Mice (n=10) were dosed BW @ 100µg (i.v.). Two-way ANOVA analysi showing significant reduction in tumor volume compared to vehicle control by i129G1, remaining significant when compared to 129hIgG1, confirming its improved direct cell killing activity.
- 129mIgG1 displays a very restricted normal tissue distribution, with weak to moderate binding of a small percentage of cells in gallbladder, ileum, liver, oesophagus, pancreas and thyroid on the AMSbio array. 129mIgG1 binds to 74% (135/182) of pancreatic tumors, 50% (46/92) of gastric tumors and 36% (100/281) of colorectal tumors.

88mIgG3 CRC binding induces ATP and HMGB1 release

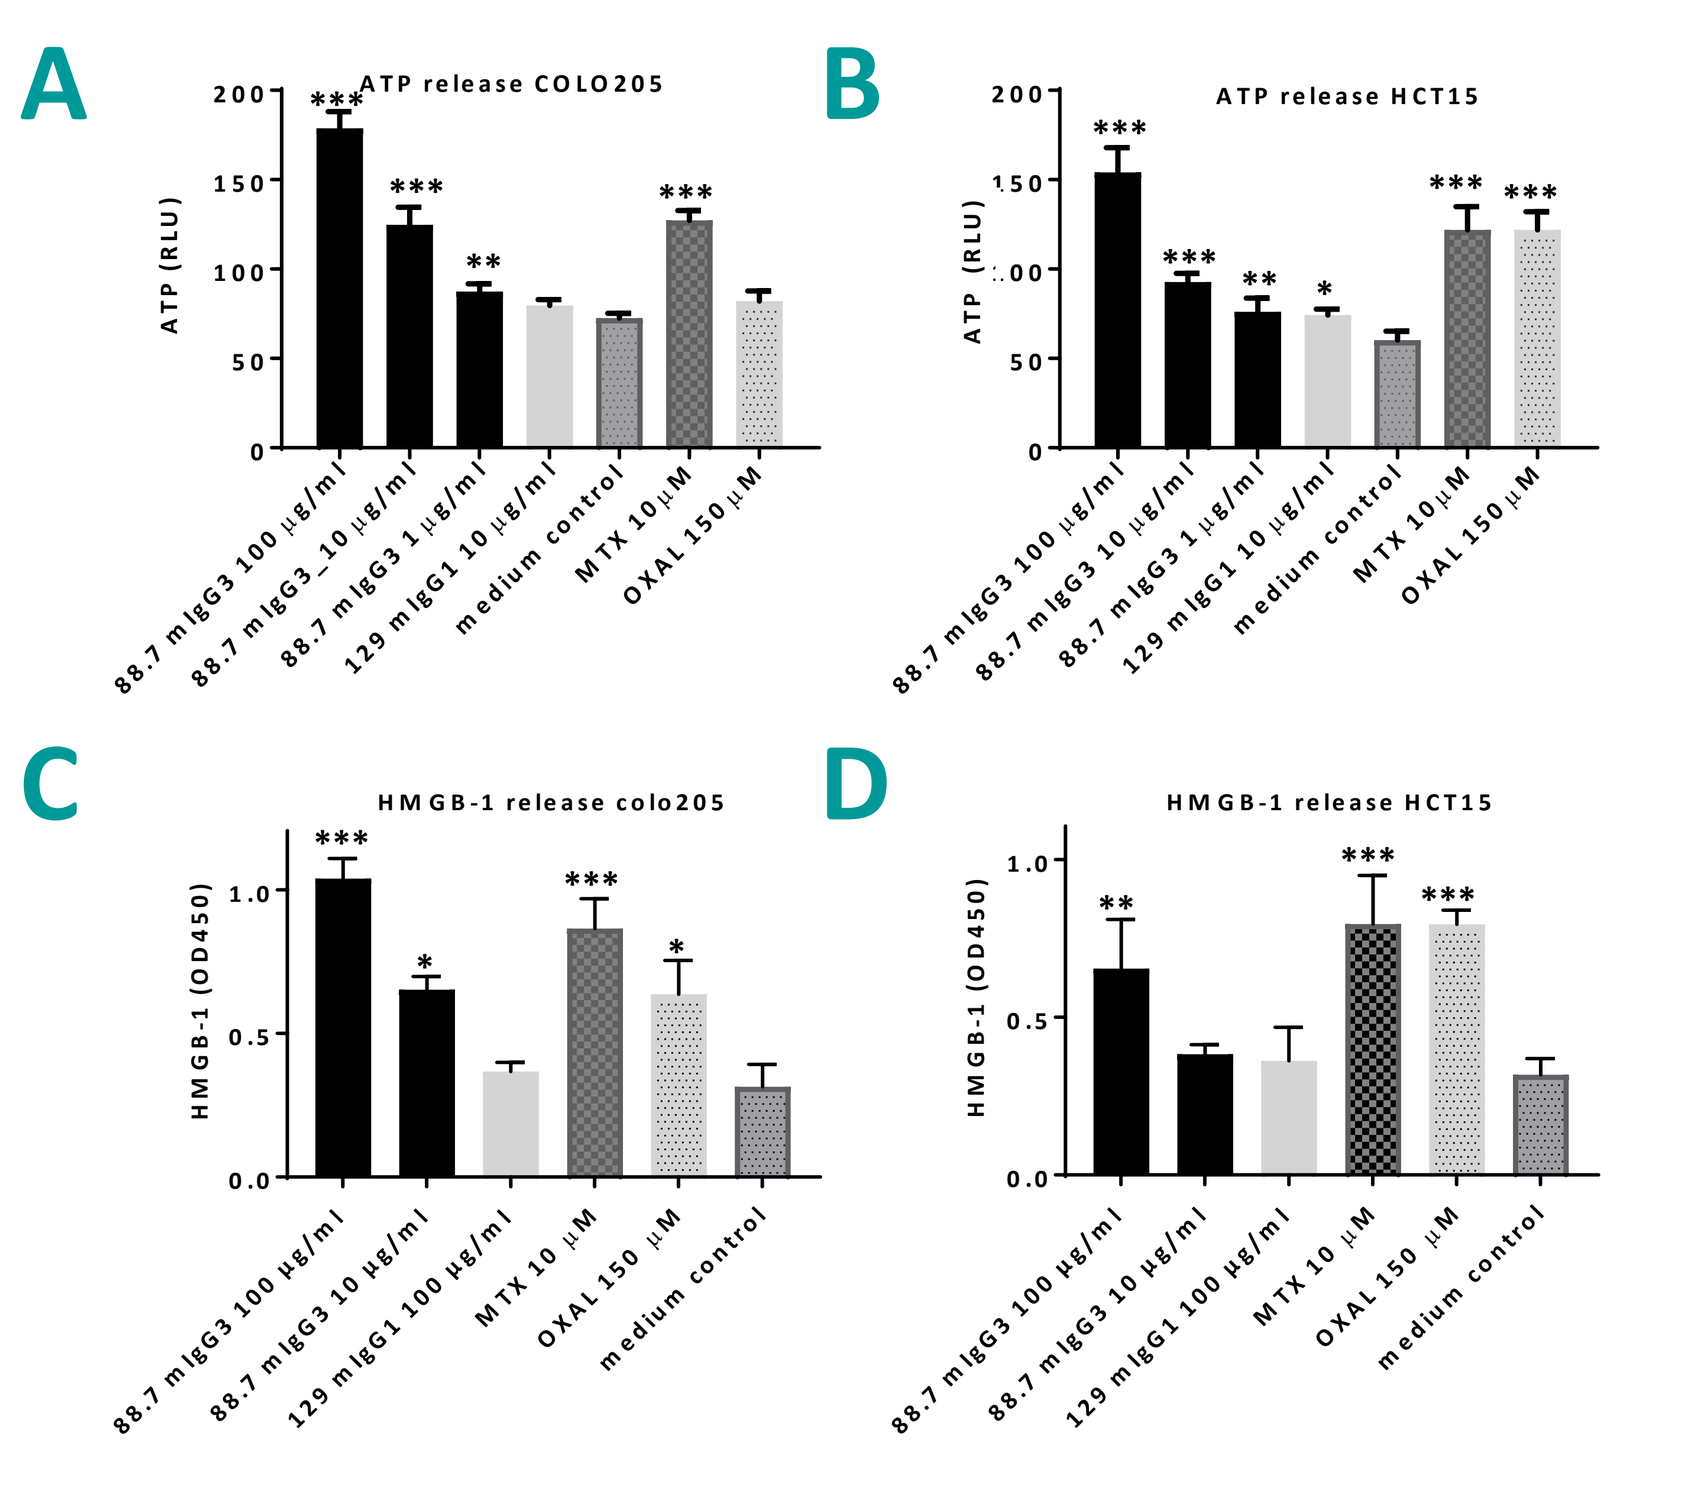
A
ATP release COLO205
ATP (RLU)
B
ATP release HCT15
C
HMGB-1 release colo205
HMGB-1 (OD450)
D
HMGB-1 release HCT15

Figure 3.

88mIgG3-treated, but not the control (129mIgG1)-treated, COLO205 and HCT15 cells release significant amounts of DAMP (danger associated molecular pattern) molecules: ATP (A,B: 8-hr treatment) and HMGB1 (C,D: 48-hr treatment), in a dose-dependent manner. The anthracyclin, mitoxantrone (MTX) was included as a positive control

DAMPs released by 88mIgG3 treatment activate innate immune cells

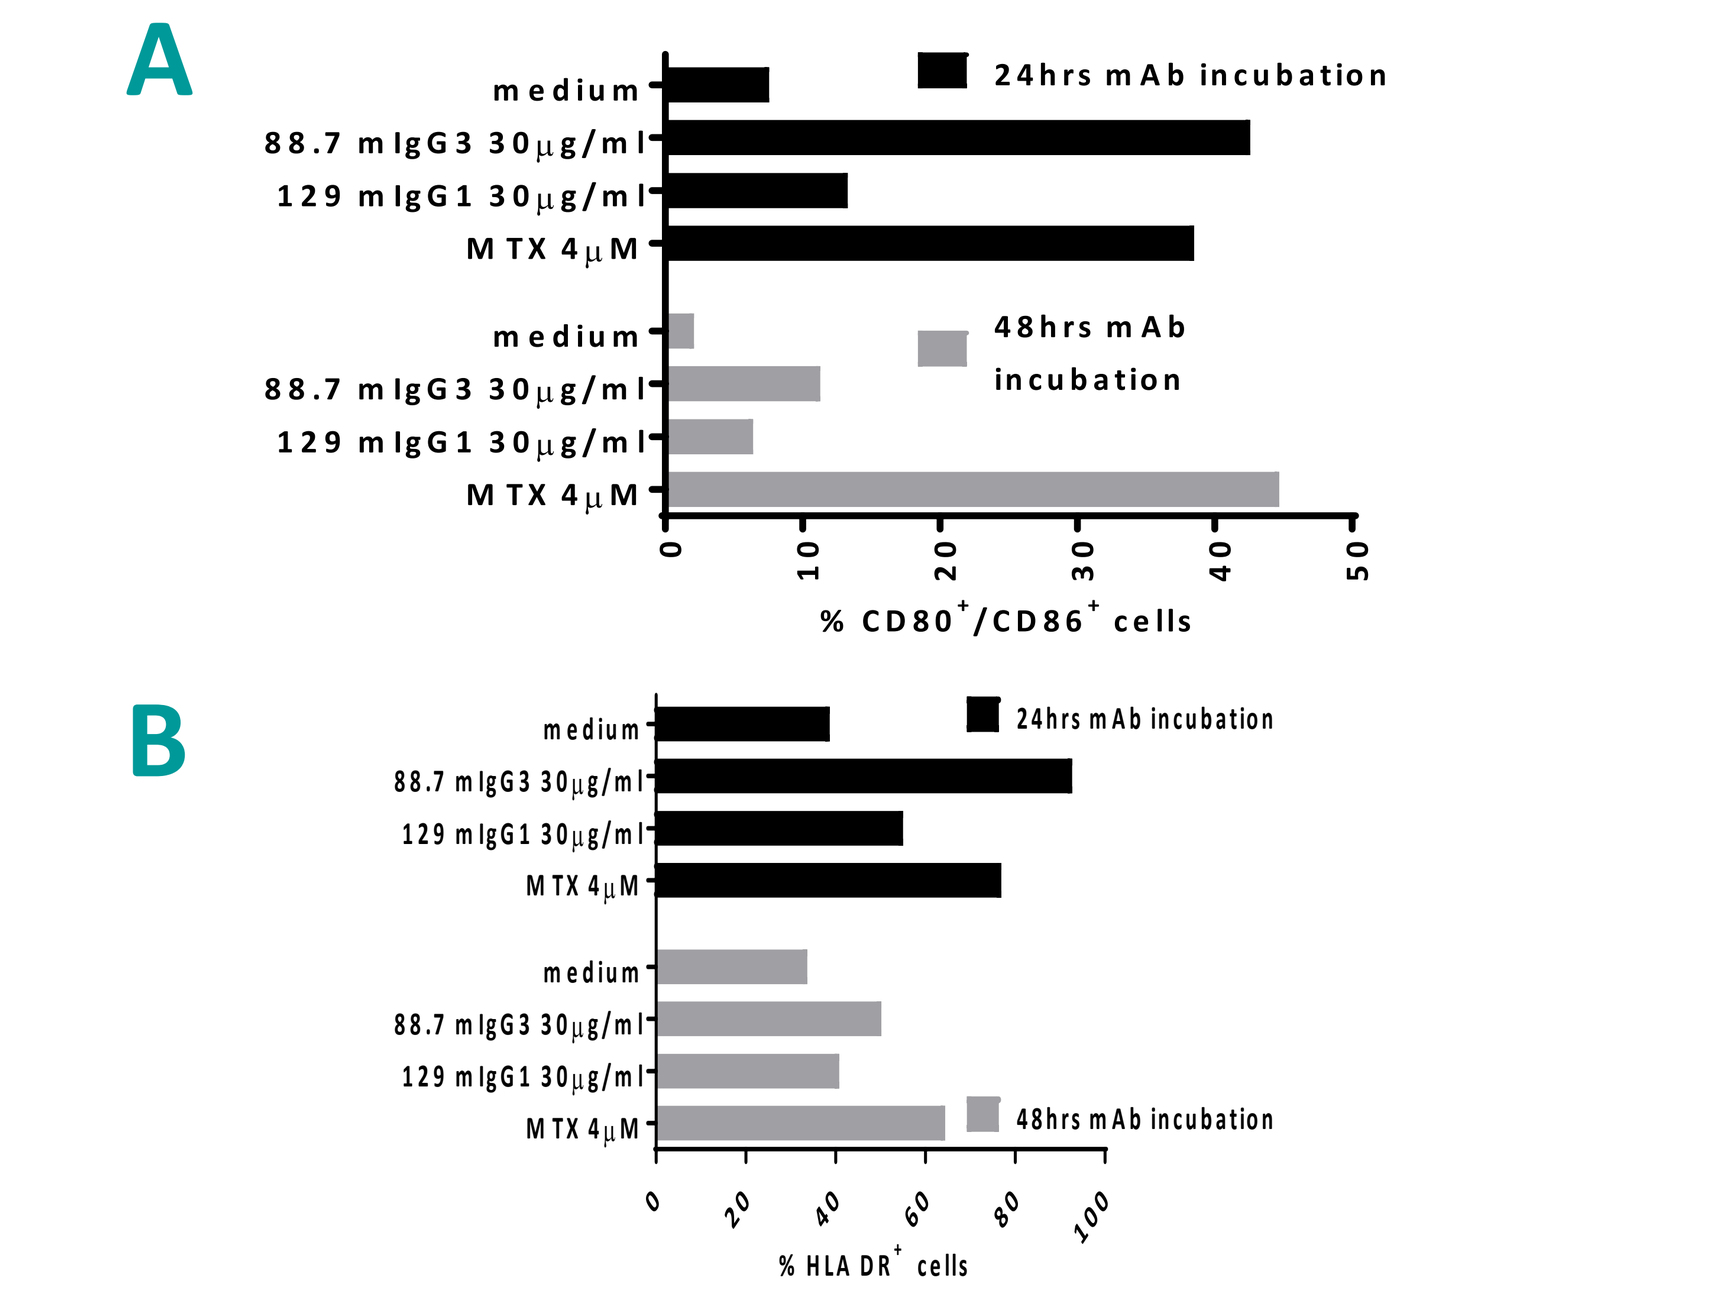
A
24hrs mAb incubation
48hrs mAb incubation
medium
88.7 mIgG3 30µg/ml
129 mIgG1 30µg/ml
MTX 4µM
% CD80+/CD86+ cells
B
% HLA DR+ cells

Figure 4.

upregulation of co-stimulatory molecules (CD80/CD86, A) and MHCII (HLA-DR, B) by immature dendritic cells in cocultures (24hr) with 88mIgG3- and MTX-treated COLO205 (24hr and 48hr). Representative results from five donors are shown. MTX, positive control; 129mIgG1 negative control

mAb engineering creates a direct cell killing 'i88G1' via transfer of selected mIgG3 residues into 88hIgG1

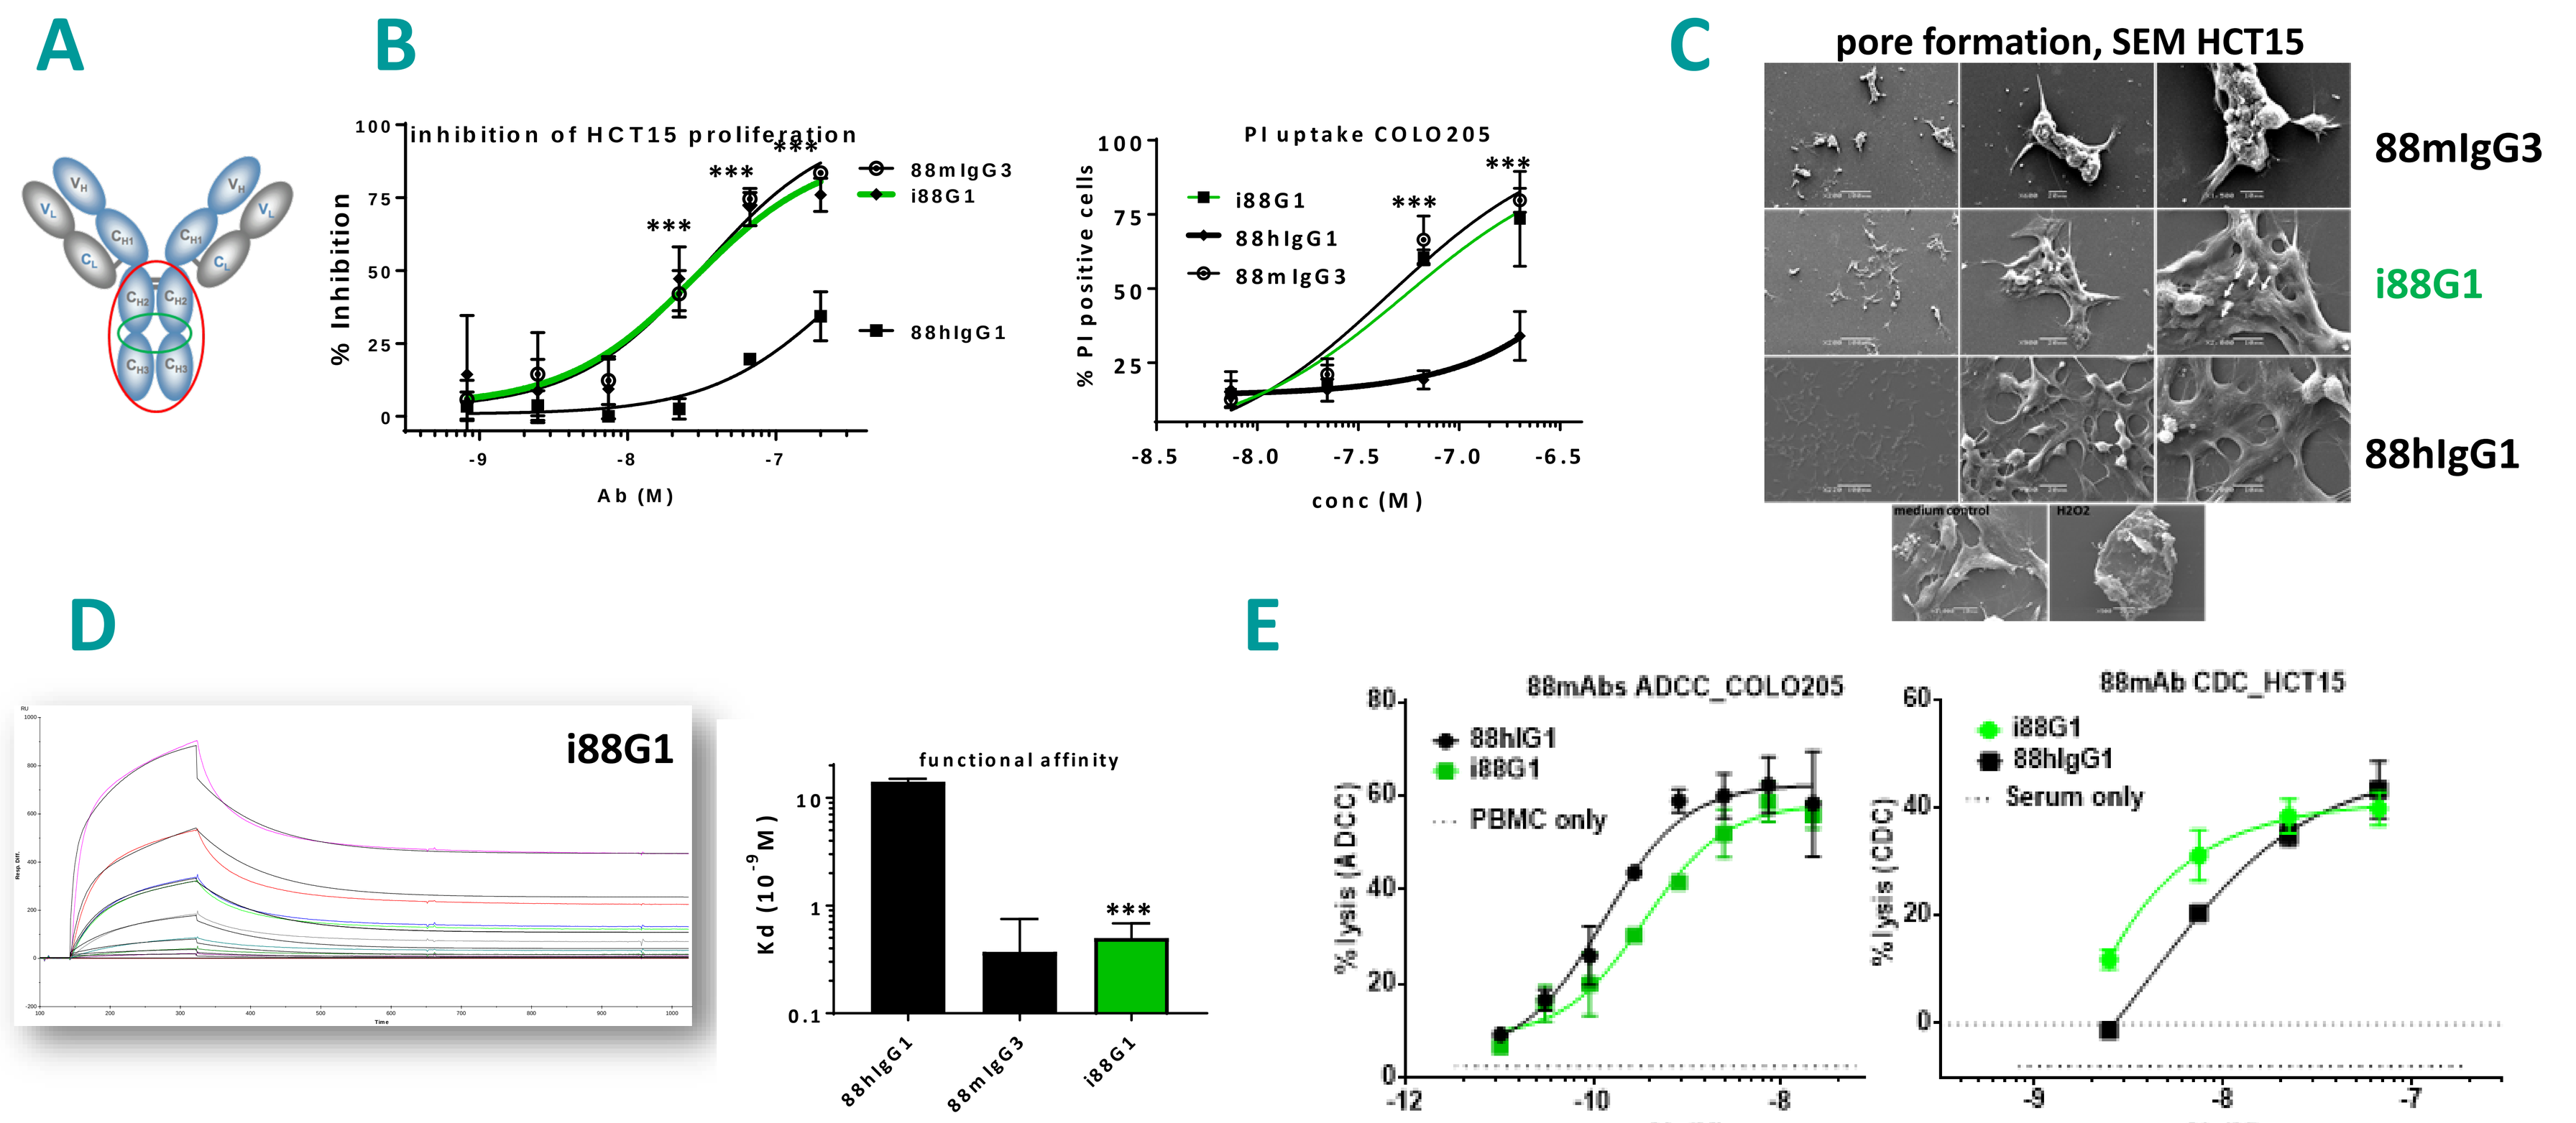
A
B
inhibition of HCT15 proliferation
88mIgG3
i88G1
88hIgG1
% Inhibition
Ab (M)
PI uptake COLO205
% PI positive cells
conc (M)
C
pore formation, SEM HCT15
88mIgG3
i88G1
88hIgG1
D
i88G1
functional affinity
Kd (10-9 M)
E
88mAbs ADCC_COLO205
PBMC only
%lysis (ADCC)
88mAb CDC_HCT15
Serum only
%lysis (CDC)
Ab (M)

Figure 5.

i88G1 contains selected constant region mIgG3 residues (A). i88G1 has significantly improved direct cell killing on COLO205 and HCT15, compared to 88hIgG1, now matching 88mIgG3 killing (B); i88G1 induces cellular aggregation and pore formation on HCT15 (C); i88G1 has significantly improved functional affinity as a result of a decreased off-rate compared to 88hIgG1 (D); i88G1 has equivalent ADCC (COLO205) and slightly improved CDC activity (HCT15) compared to 88hIgG1 (E)

references

[1] Chua JX, Vankemmelbeke M, McIntosh RS, Clarke PA, Moss R, Parsons T and Durrant LG. **Clinical Cancer Research 2015** Jul 1;21(13):2963-74
[2] Greenspan NS and Cooper LJ. **Springer Semin Immunopathol 1993**;15(2-3):275-91
[3]Kroemer G, Galluzzi L, Kepp O, and Zitvogel L. **Annual Review of Immunology 2013**;31: 51-72

PRINTED BY SCIENCEPOSTERS .co.uk